Direct and Selective Myosin-Inhibition Stabilizes the Super-Relaxed State of Myosin and Blunts Post-Tetanic Potentiation in Fast-Skeletal Muscle

Newell-Stamper BL[1]*, Lehman S[1]*, DuVall M[1], Madden M[1], Evanchik M[1], Qian Y[1], Barthel B[1], Russell AJ[1†], del Rio CL[1†]

[1]*Edgewise Therapeutics, BioFrontiers Institute, University of Colorado, Boulder, CO 80303, USA.*

Post-tetanic potentiation (PTP) describes the acute recruitment of force that follows repetitive stimulation of striated muscle. The progressive destabilization/recruitment of myosin-heads from the thick-filament backbone, and their subsequent incomplete recovery, has been postulated as a potential mechanism for PTP in fast-twitch skeletal muscles. Paradoxical PTP enhancements have been described in various models of muscular dystrophy, suggesting that contraction-induced force amplification and thick-filament disorder play a role in the pathophysiology of injury. These studies leveraged a selective fast-skeletal myosin modulator, EDG-4131, to test the hypothesis that PTP is mediated by the population of myosin-heads available to form cross-bridges and the release of myosin-heads from the reserve super-relaxed-state (SRX)

METHODS

- **Using a NADH-coupled regenerative assay, the ATPase activity of purified rabbit fast skeletal (psoas) myofibrils, porcine ventricular myofibrils, and fast skeletal (psoas) chymotrypsin-digested myosin sub-fragment 1 (S1) was measured. EDG-4131 was incubated in each tissue type (0-100µM). Curves were fit with Michaelis-Menten kinetics to calculate the maximum ATPase rate (mAmp/min) and EDG-4131 activity levels were normalized to DMSO activity. Potency of EDG-4131 on inhibition of activity (IC-50) was calculated for each tissue type.**
- **SRX/DRX ratio was measured in tissue-purified rabbit skeletal psoas heavy meromyosin (HMM) using a modified mant-ATP based ATPase assay (adapted from [1,2]). Mant-ATP (0.4µM) was mixed with HMM (0.4µM), aged for 60 seconds, then rapidly mixed with unlabeled ATP (16mM). Mant-ATP fluorescence decay was monitored at 385nm at 1Hz for 500 seconds. Data were fit with a double-exponential decay to calculate %DRX population (%SRX=100-%DRX) and ATPase rates for myosin heads in DRX and SRX states (KDRX and KSRX, respectively) (example curve shown to right). Simulated curves (100% DRX and 100% SRX) demonstrate the presence of both DRX and SRX myosin heads in rabbit psoas HMM.**

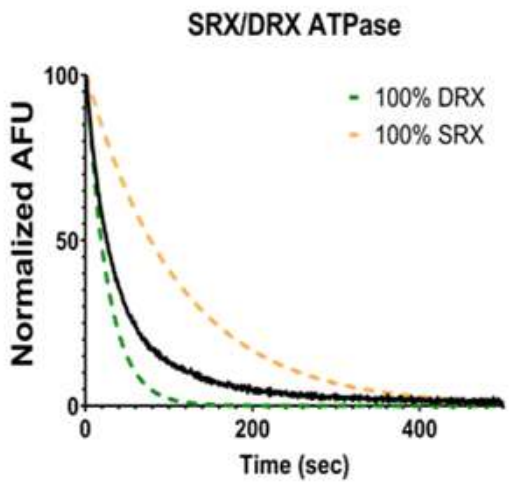
SRX/DRX ATPase
100% DRX
100% SRX
Normalized AFU
Time (sec)

- **Ex vivo muscle force was measured using the mouse fast-twitch extensor digitorum longus (EDL) muscle. The muscle was isolated from C57BL/6 mice under isoflurane and was put into oxygenated Krebs Ringer Mammalian Buffer at 25°C where it equilibrated for 10 minutes. All experiments were approved by the CU Boulder IACUC.**
- **Force potentiation was measured using stimulation frequencies 1-15 Hz (adapted from [3]). EDG-4131 (2.5 µM) was added to the muscle bath after a baseline was measured at each stimulation frequency. After a 10 minute incubation the next series of stimulations were applied. The 1 Hz frequency produced the largest potentiation response and was used to assess the effect of compound. Force inhibition was assessed by comparing twitch and tetanus forces before/after addition of EDG-4131.**

RESULTS

EDG-4131 selectively inhibits fast skeletal myosin ATPase activity.

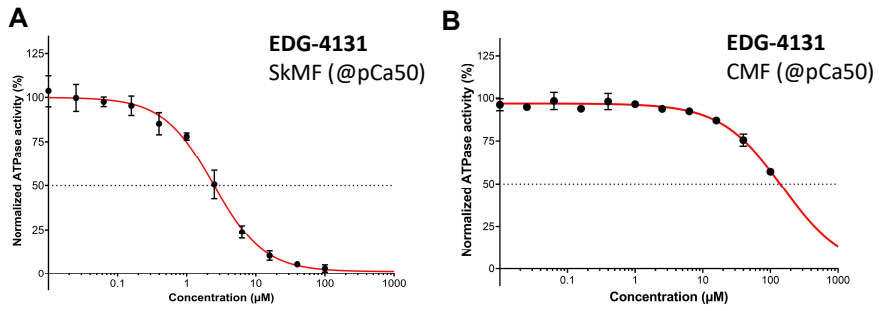
A
EDG-4131
SkMF (@pCa50)
B
EDG-4131
CMF (@pCa50)
Normalized ATPase activity (%)
Concentration (µM)

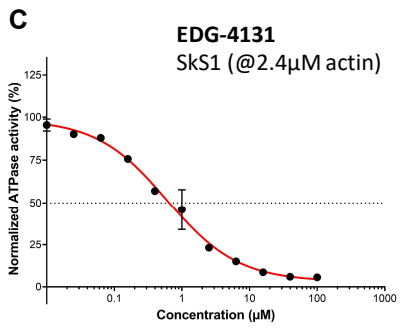
C
EDG-4131
SkS1 (@2.4µM actin)
Normalized ATPase activity (%)
Concentration (µM)

ATPase activity was dose-dependently inhibited in rabbit skeletal myofibrils (skMF) (A, table) and purified rabbit psoas myosin S1 (skS1) (C, table) with EDG-4131, but minimal inhibition was seen in porcine ventricular myofibrils (cMF) (B, table). Data reported as average ± SEM.

Tissue Source	IC_{50} (µM)	ATPase @ 2.5µM (%)
skMF (fast)	1.06	52.2 ± 3.2
cMF (cardiac)	>100	93.7 ± 1.0
skS1 (fast)	0.65	23.2 ± 1.0

EDG-4131 stabilizes the super relaxed (SRX) state of myosin in rabbit psoas HMM.

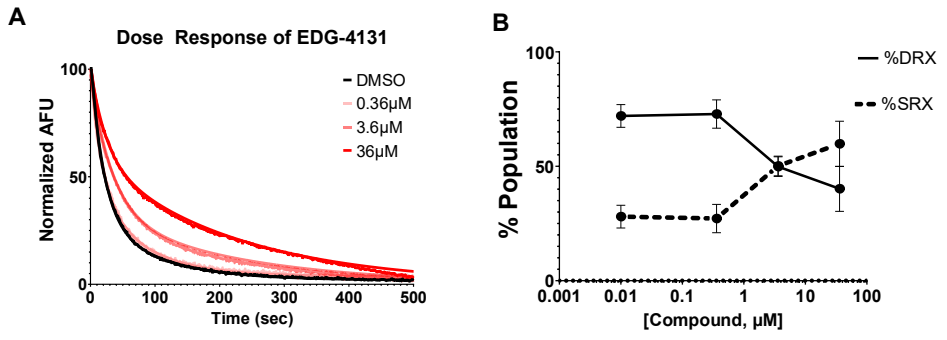
A
Dose Response of EDG-4131
DMSO
0.36µM
3.6µM
36µM
Normalized AFU
Time (sec)
B
%DRX
%SRX
% Population
[Compound, µM]

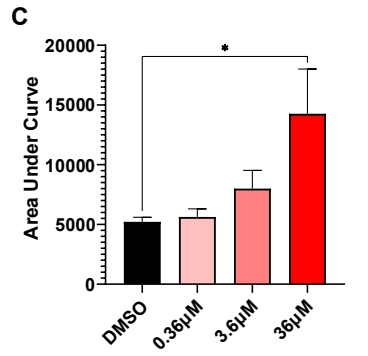
C
Area Under Curve
DMSO
0.36µM
3.6µM
36µM

The population of myosin heads in the SRX state increased with EDG-4131, suggesting stabilization of the myosin OFF-state (A & B, table). EDG-4131 treatment increased the area under the curve, suggesting decreased resting ATP consumption with compound (C). Data reported as average ± SEM. *p= 0.03 vs EDG-4131 treatment, One-way ANOVA

	DMSO	0.36 µM	3.6 µM	36 µM
%DRX	74.4 ± 4.5	72.8 ± 6.2	49.0 ± 4.3	37.9 ± 11.8*
%SRX	25.5 ± 4.5	27.2 ± 6.2	50.1 ± 4.3	62.1 ± 11.8*
K_{DRX} (ATP/s)	0.06 ± 0.016	0.071 ± 0.03	0.046 ± 0.015	0.068 ± 0.02
K_{SRX} (ATP/s)	0.012 ± 0.002	0.011 ± 0.005	0.009 ± 0.004	0.010 ± 0.004

EDG-4131 attenuates twitch and tetanic force

EDG-4131 inhibits potentiation in fast skeletal muscle

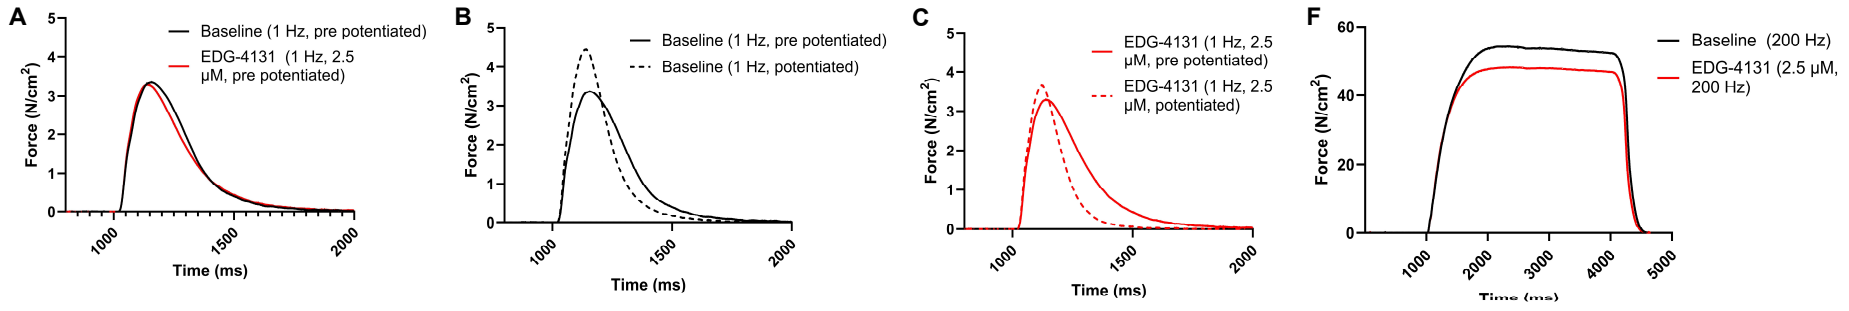
A
Baseline (1 Hz, pre potentiated)
EDG-4131 (1 Hz, 2.5 µM, pre potentiated)
B
Baseline (1 Hz, pre potentiated)
Baseline (1 Hz, potentiated)
C
EDG-4131 (1 Hz, 2.5 µM, pre potentiated)
EDG-4131 (1 Hz, 2.5 µM, potentiated)
F
Baseline (200 Hz)
EDG-4131 (2.5 µM, 200 Hz)
Force (N/cm²)
Time (ms)

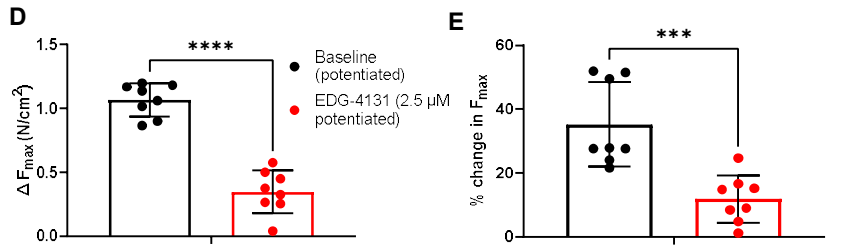
D

Baseline (potentiated)
EDG-4131 (2.5 µM potentiated)
E

	Baseline (pre-)	Baseline (potentiated)	EDG-4131 (pre-)	EDG-4131 (potentiated)
Fmax (N/cm²)	3.36 ± .008	4.43 ± .005	3.30 ± 0.007	3.66 ± .004
Δ Fmax	--	1.07 ± .046	--	0.35 ± .059*
% change	--	35.19 ± 4.68	--	11.81 ± 2.65*

A) After a 10-min. incubation with EDG-4131 pre-potentiation force was not reduced. B) Potentiation at baseline resulted in a 35.2% ± 4.68 SEM increase in force. C) Potentiation after a 10-min incubation with EDG-4131 resulted in a 11.81% ± 2.65 SEM increase in force. D and E) EDG-4131 blunted Fmax potentiation in both absolute (a +0.35 ± .059 vs. +1.07 ± .046 N/cm² increase at baseline, P < 0.0001, table) or relative terms (+11.8 ± 2.7 vs. 35.2 ± 4.7 % at baseline, P < 0.001, table). F) Tetanic force measured late in the protocol was attenuated with EDG-4131 by 6.203 N/cm² ± 1.81 SEM (11.26 % ± 3.4 SEM) on average.

Potentiation protocol

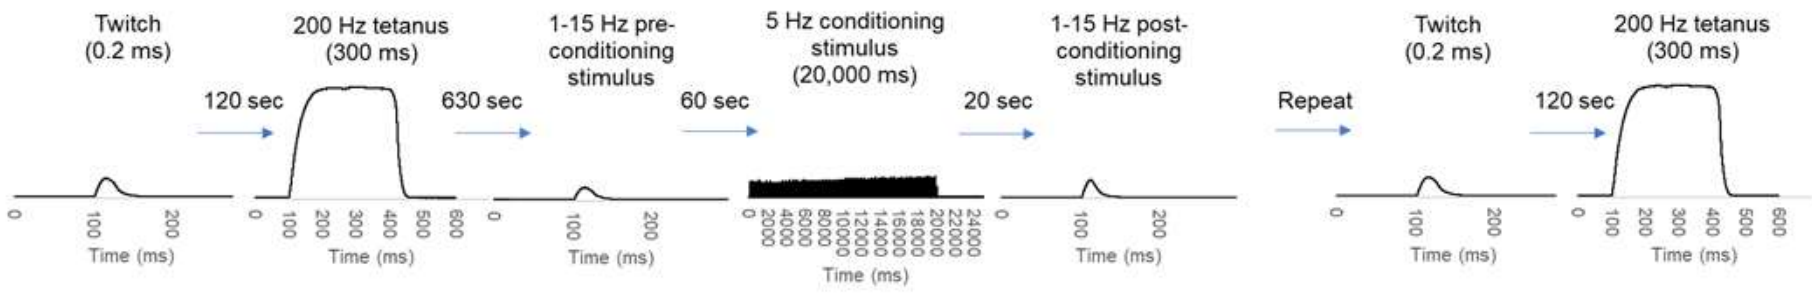
Twitch (0.2 ms)
200 Hz tetanus (300 ms)
1-15 Hz pre-conditioning stimulus
5 Hz conditioning stimulus (20,000 ms)
1-15 Hz post-conditioning stimulus
Twitch (0.2 ms)
200 Hz tetanus (300 ms)
120 sec
630 sec
60 sec
20 sec
Repeat
120 sec
Time (ms)

DISCUSSION

- EDG-4131 is a novel myosin inhibitor that selectively reduces the ATPase activity of fast skeletal myosin in both myofibrils and myosin-S1.
- EDG-4131 stabilizes the population of myosin heads in the ultra-low ATP consumption SRX state, decreasing the number of heads available to form actomyosin force-generating cross-bridges.
- The mouse EDL, a fast-skeletal muscle, shows strong potentiation with ~30% force-enhancements post-potentiation.
- Pharmacological stabilization of the myosin's SRX state with EDG-4131 blunted potentiation in EDL muscles, with negligible impact in pre-potentiated twitches. Twitch and tetanus force was attenuated with EDG-4131.
- These data demonstrate a mechanistic link between the de-stabilization of myosin heads in the thick-filament and recruitment of force via potentiation in fast-skeletal muscle.

Pharmacological stabilization of the myosin's SRX state with EDG-4131, a selective myosin-modulator, blunted PTP in EDL muscles, with negligible impact in pre-potentiated twitches.

REFERENCES

1. Anderson, R.et al. Deciphering the super relaxed state of human β-cardiac myosin and the mode of action of mavacamten from myosin molecules to muscle fibers. Proc NatlAcad Sci U S A115, E8143-E8152, doi:10.1073/pnas.1809540115 (2018).
2. Gollapudi, S., Yu, M., Gan, Q. & Nag, S. Synthetic thick filaments: A new avenue for better understanding the myosin super-relaxed state in healthy, diseased, and mavacamten-treated cardiac systems.J Biol Chem296, 100114,doi:10.1074/jbc.RA120.016506 (2021).
3. Vandenboom, R., Grange, RW., Houston, ME. Threshold for force potentiation associated with skeletal myosin phosphorylation. Am J Physiol. 1993 Dec;265(6 Pt 1):C1456-62. doi: 10.1152/ajpcell.1993.265.6.C1456. PMID: 8279509.

COMPETING INTERESTS: BN-S, SL, MD, MM, ME, CLdR, YQ, BB, and AR are employees of Edgewise Therapeutics and hold financial interests (stock and/or stock options).

†:Please contact either AJR (arussell@edgewisetx.com), CLdR (cdelrio@edgewisetx.com) or BN-S (bstamper@edgewisetx.com).