A Phase 1, Randomized, Double-blind, Placebo-controlled, Single and Multiple Ascending Dose Study of the Safety, Tolerability, Pharmacokinetics, and Pharmacodynamics of EDG-7500 in Healthy Adults

Marc Semigran MD, Christopher Dufton PhD, Daniel Gretler MD, Marc Evanchik MS

Edgewise Therapeutics, Boulder CO

Background

Hypertrophic cardiomyopathy (HCM) is a chronic, progressive disease of the sarcomere. A significant portion of affected individuals have at least one identifiable mutation in a gene that encodes sarcomere proteins. Regardless of the genetic variant present, excess myosin-actin crossbridge formation in systole and diastole leads to hyperdynamic contraction and impaired relaxation. Over time these abnormalities lead to tissue remodeling characterized histologically by myocyte hypertrophy, myofilament disarray, microvascular remodeling, and fibrosis. Clinically, patients experience fatigue, exertional dyspnea, and an increased risk of sudden cardiac death.

Two thirds of patients with HCM have obstruction of outflow from the heart due to apposition of the anterior leaflet of the mitral valve with the interventricular septum at the left ventricular outflow tract (LVOT). It has been observed that systolic anterior motion results from abnormalities in early systolic blood flow impacting the posterior surface of the protruding mitral value leaflet.[1] Relief of LVOT obstruction, either via surgical, interventional, or pharmacologic therapies has been associated with improved exercise capacity in obstructive HCM patients.

EDG-7500 is a first-in-class, oral, selective, cardiac sarcomere modulator, specifically designed to slow early contraction velocity and address impaired cardiac relaxation associated with HCM and other diseases of diastolic dysfunction. This molecule is initially being developed for the treatment of HCM. Preclinical models demonstrate improved left ventricular compliance and distensibility and ameliorate hyperdynamic systolic contraction and LVOT obstruction.

Overall Program Hypothesis

EDG-7500 will slow the rate of early LV contraction and speed the rate of relaxation, while avoiding an undesired excessive drop in systolic performance.

Study Objectives

- The purpose of this first-in-human study is to evaluate the initial safety, tolerability, pharmacokinetics (PK), pharmacodynamics (PD), food effect (FE), and relative bioavailability (rBA) of suspension and solid formulations of EDG-7500 in healthy adults.
- Information derived from this study will guide further clinical development of EDG-7500 as a potential therapy for patients with HCM and other diseases of diastolic dysfunction.

Key Eligibility Criteria

- Healthy non-pregnant females or males
- Age ≥ 18 to < 72 years
- No clinically significant cardiac structural abnormalities
- Left ventricular ejection fraction (LVEF) ≥ 0.60

Study Protocol (First Cohort enrolled: September 2023)

- This is a 3-part, first-in-human, double-blind (Sponsor unblinded), randomized, placebo-controlled study conducted at a single site in the US. The study will enroll approximately 126 subjects into single ascending dose (SAD), multiple ascending dose (MAD), and FE/rBA cohorts.
 - SAD: Double-blind (Sponsor unblinded), randomized, placebo-controlled, sequential group study in approximately 48 healthy adult subjects.
 - MAD: Double-blind (Sponsor unblinded), randomized, placebo-controlled, sequential group study in up to 50 healthy adult subjects, ages ≥ 18 to < 72 years.
 - Food effect and solid oral dose form evaluation: randomized, open-label, 3-period, crossover study in approximately 9 to 12 healthy adults, depending on PK variability.
 - The tentative dose ascension scheme consists of 5, 15, 50, 100, 200, 300 mg. At each dose level, 2 sentinel subjects (1 EDG-7500, 1 placebo) will receive study drug at least 48 h prior to dosing of the remaining subjects.
- After the completion of each dose level, the safety data through Day 5, including routine laboratory data as well as echocardiographic data (in particular LVEF) relevant to safety will be reviewed by a Safety Review Committee (SRC) before deciding on the next dose level.
- The SRC may choose to keep the tentative dose levels shown above, lower the dose increment, decrease the dose from a previous dose level, or repeat the same dose.

Nonclinical Data[2,3]

Healthy Dog Model

EDG-7500 Slows LV Pressure Development in Early Systole

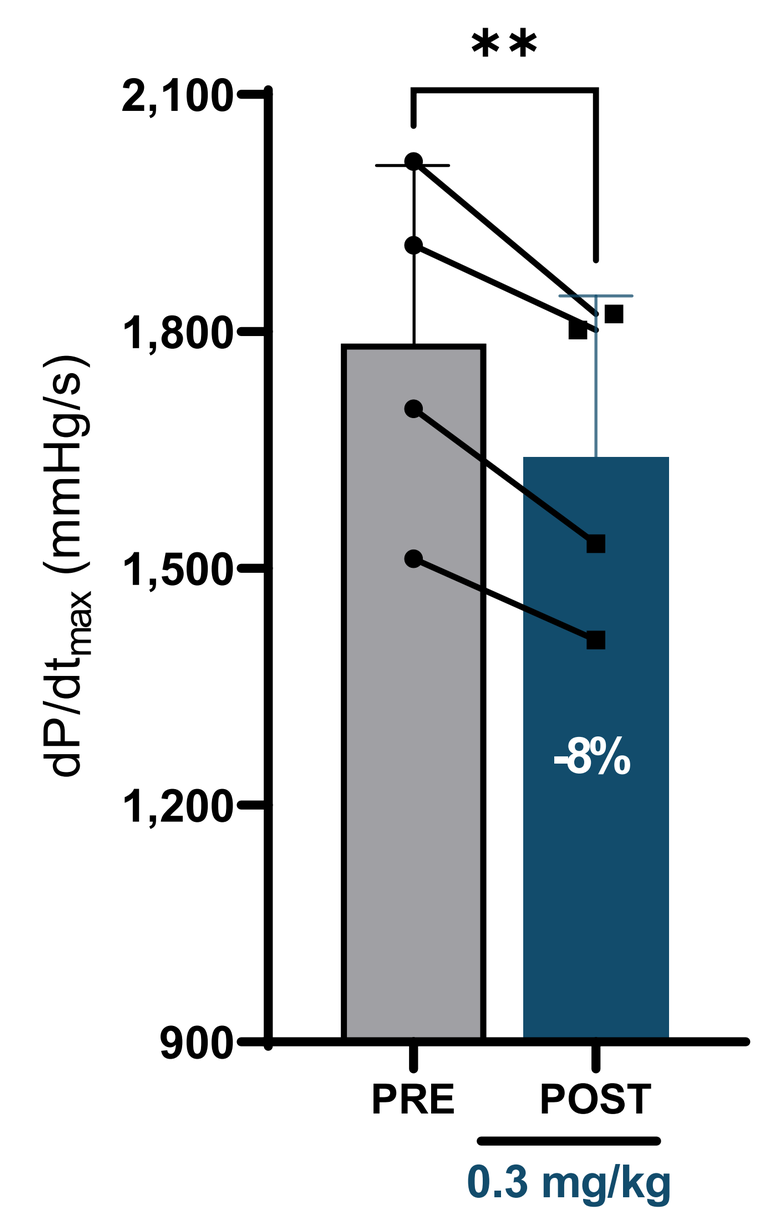
**
2,100
1,800
1,500
1,200
900
dP/dtmax (mmHg/s)
-8%
PRE
POST
0.3 mg/kg

EDG-7500 Prolongs the Pre-ejection Period

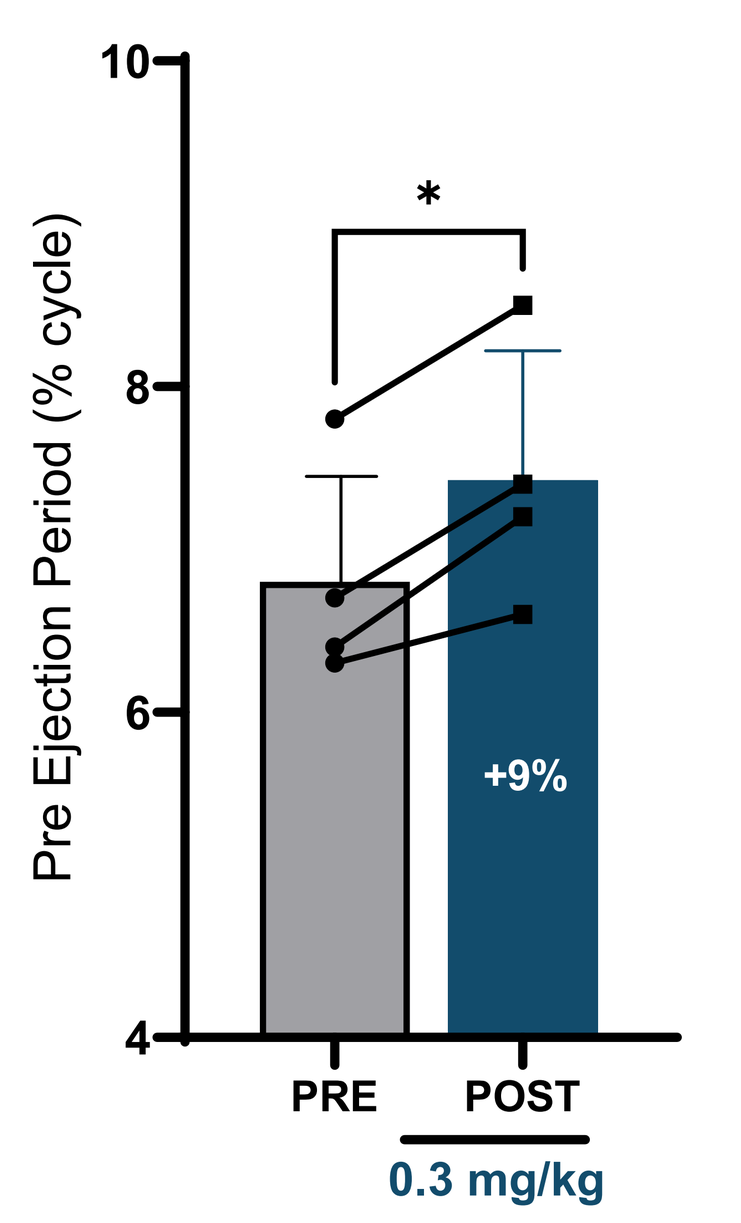
*
10
8
6
4
Pre Ejection Period (% cycle)
+9%
PRE
POST
0.3 mg/kg

SD shown; Footnotes: * $p < 0.05$; ** $p < 0.01$
Abbreviations: dP/dt_{max}, the maximum rate of rise in ventricular pressure; LV, left ventricle

EDG-7500 Increases LV Compliance
End Diastolic Volume Increased; End Diastolic Pressure Maintained

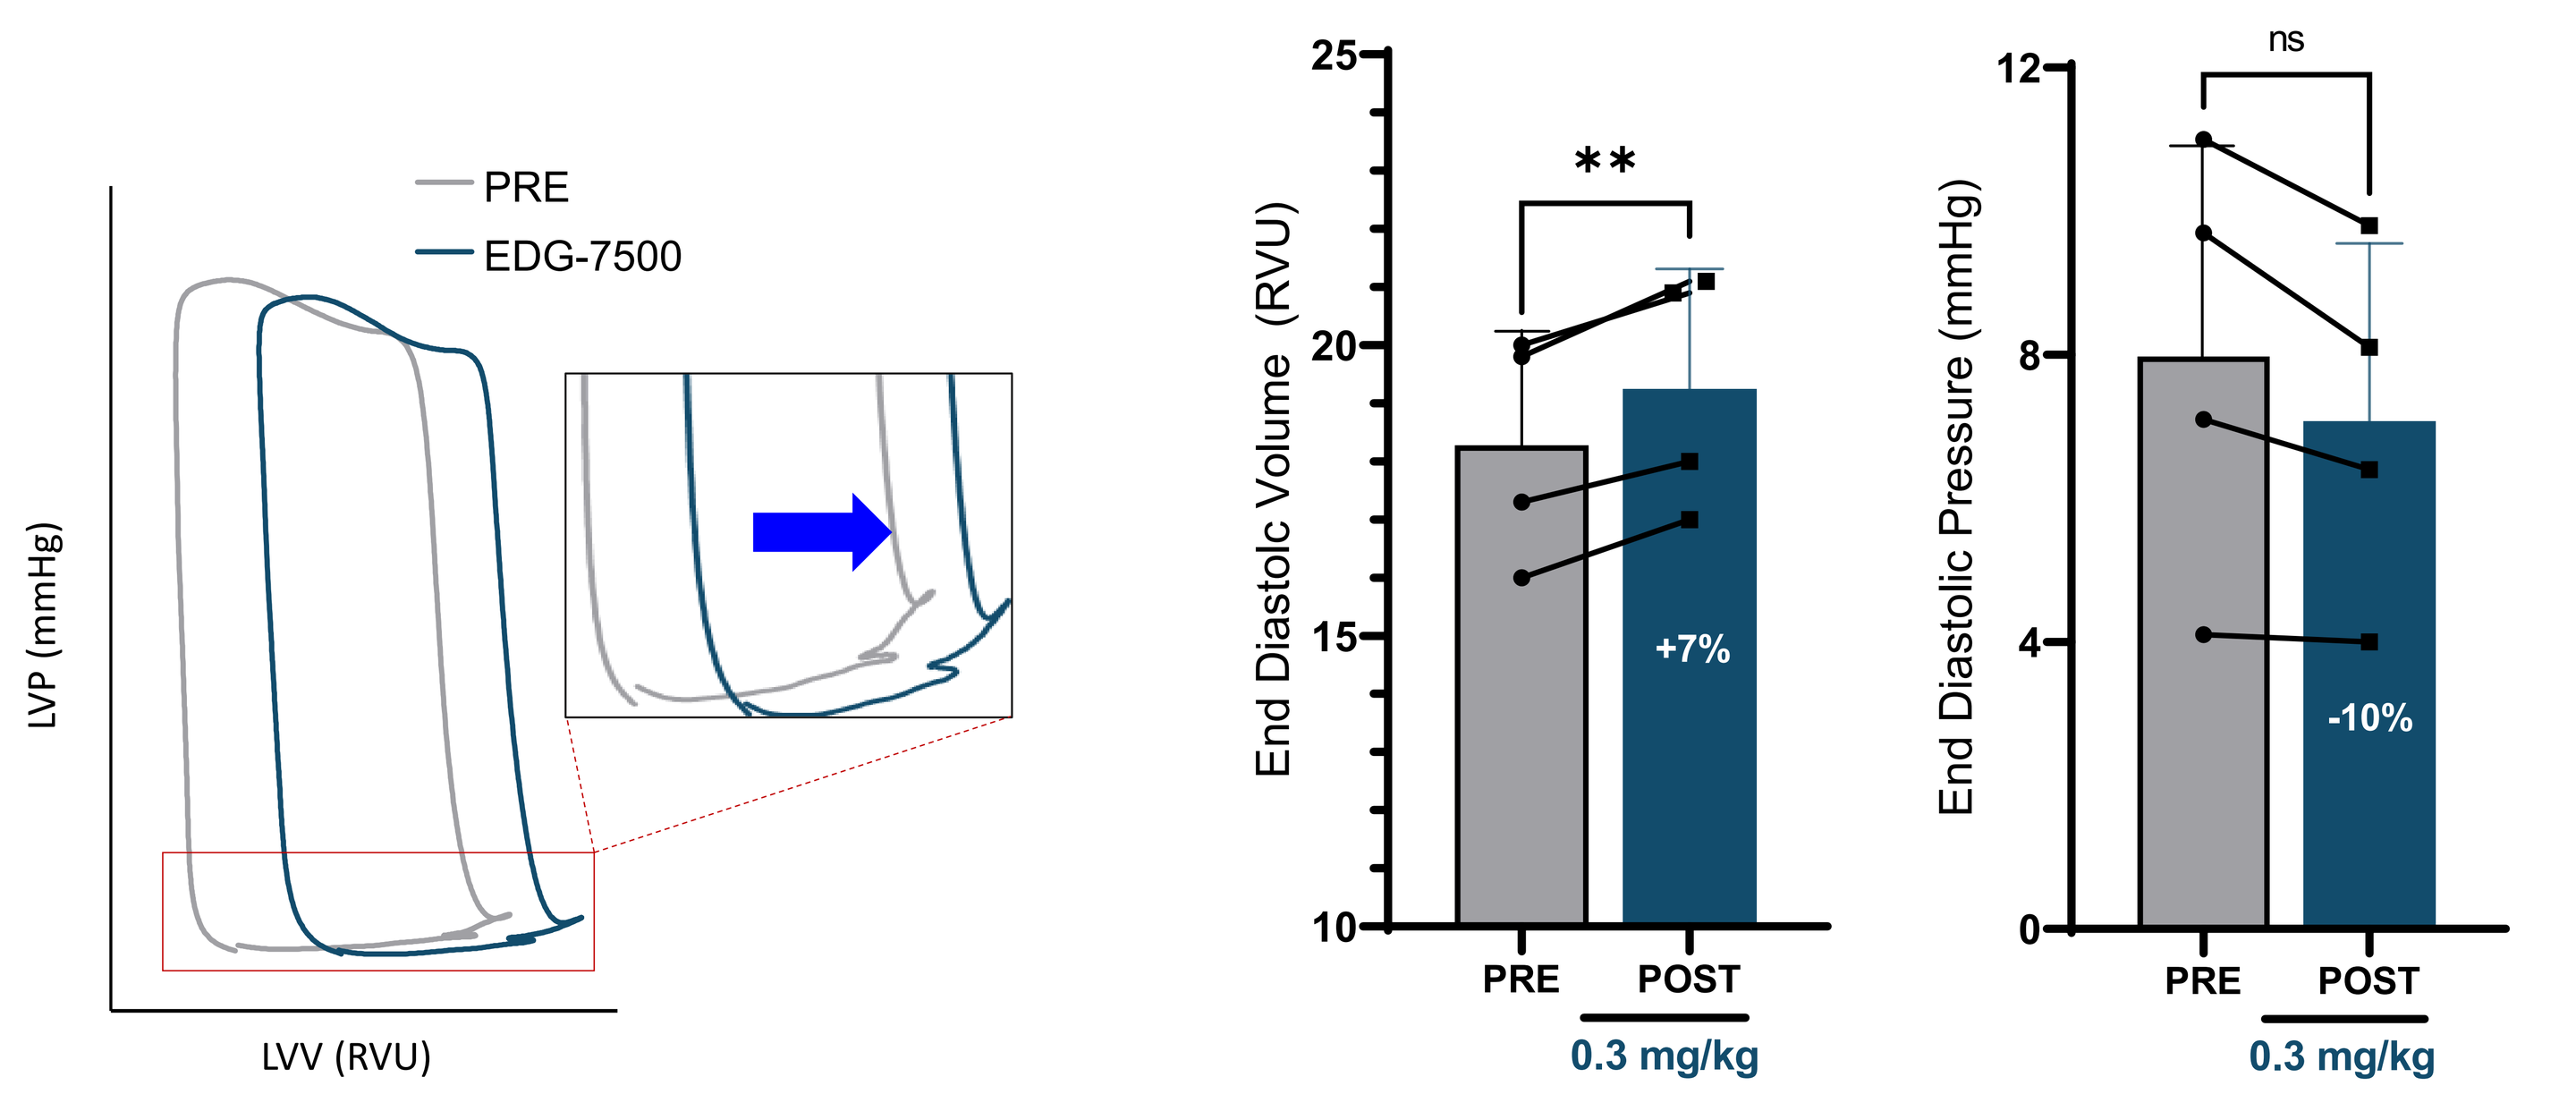
PRE
EDG-7500
LVP (mmHg)
LVV (RVU)
**
25
20
15
10
End Diastolc Volume (RVU)
+7%
PRE
POST
0.3 mg/kg
ns
12
8
4
0
End Diastolic Pressure (mmHg)
-10%
PRE
POST
0.3 mg/kg

SD shown; Footnotes: ** $p < 0.01$
Abbreviations: LVP, LV pressure; LVV, LV volume; RVU, relative volume units

A31P MyBP-C Cat Model

LVFS Maintained with Oral EDG-7500 Treatment

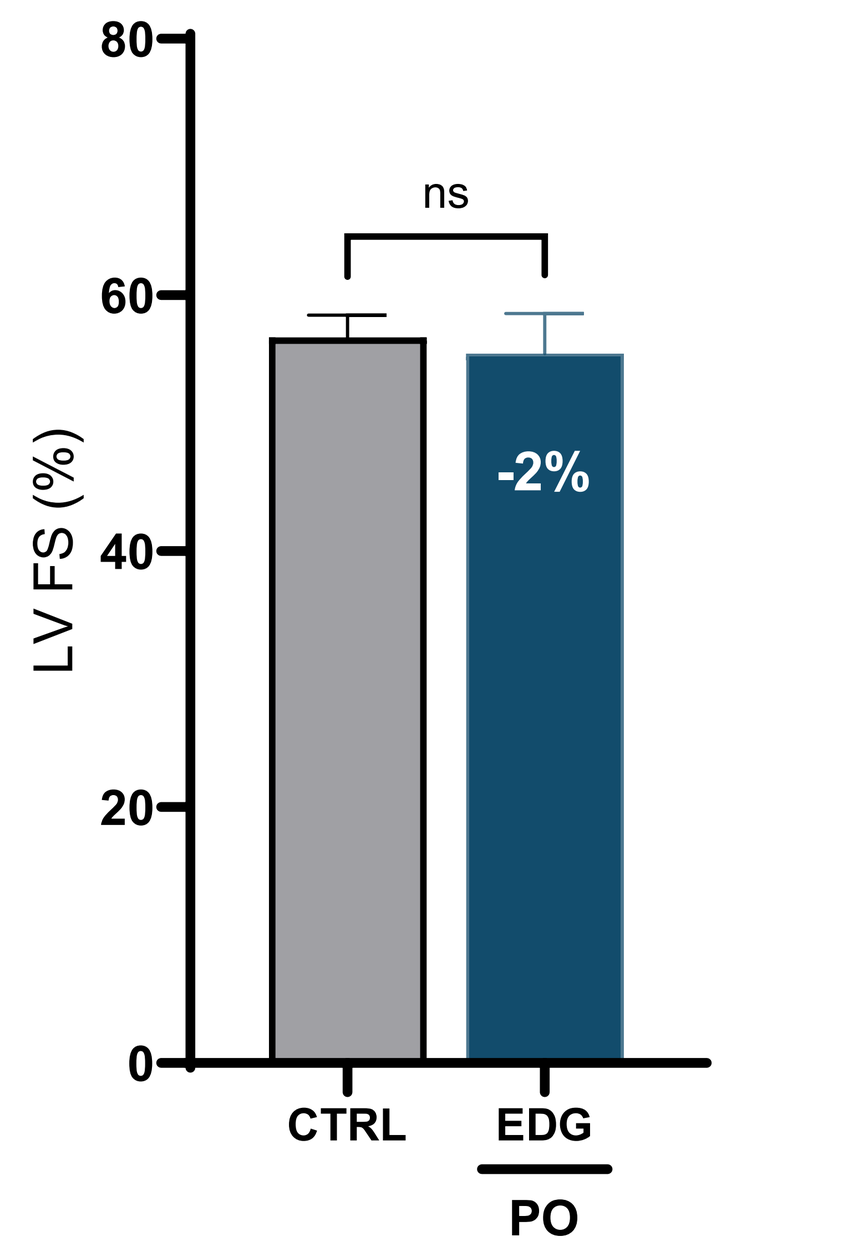
80
60
40
20
0
LV FS (%)
ns
-2%
CTRL
EDG
PO

EDG-7500 Driven LVOT Gradient Relief

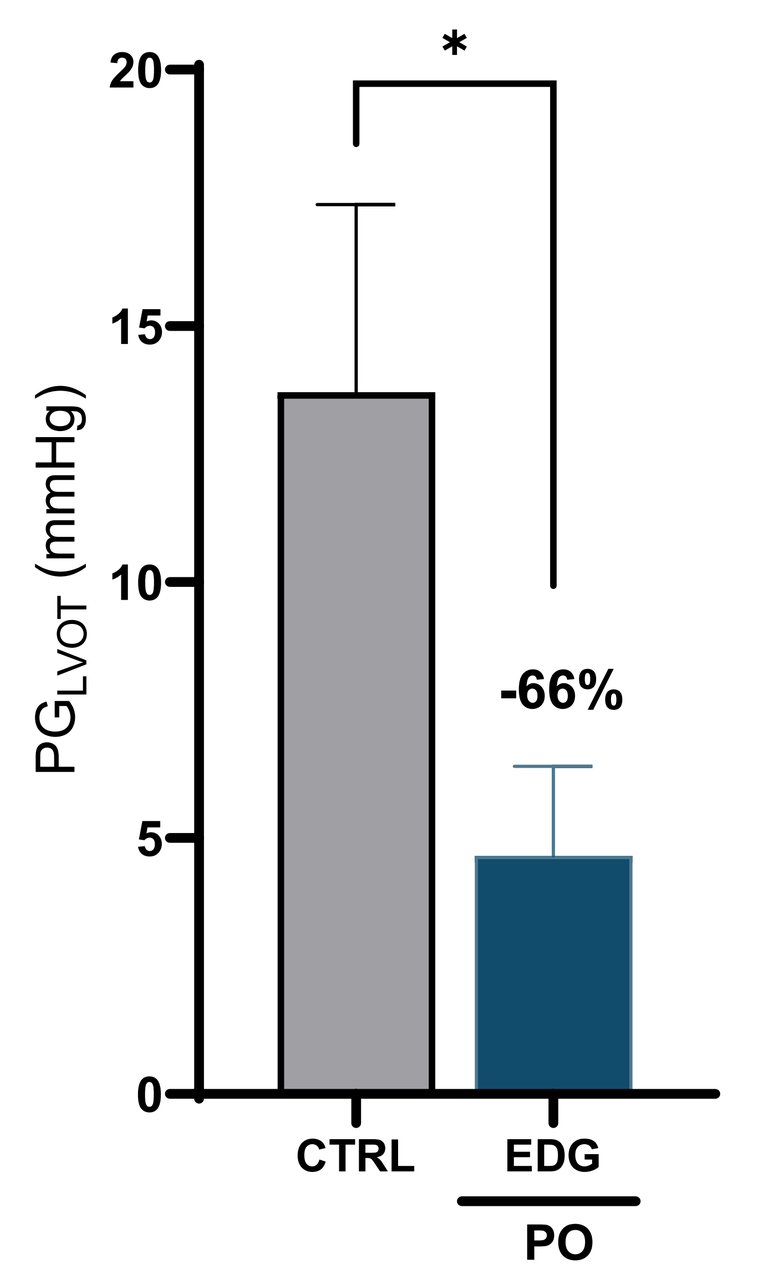
20
15
10
5
0
PGLVOT (mmHg)
*
-66%
CTRL
EDG
PO

Footnotes: * $p < 0.05$

Outcome Measures of Interest

- Safety and tolerability
- Single dose and steady state plasma and urine PK profile
- QTc interval
- LV function assessed by echocardiographic measures
- Changes in LV function from rest to submaximal exercise assessed by echocardiographic measures
- Circulating cardiac biomarkers

References

[1] Ro R et al. J Am Coll Cardiol. 2014;64:1984-95.
[2] Edgewise Therapeutics: Data on file (https://edgewisetx.com)
[3] Kaplan J, et al. J Am Coll Cardiol. 2023;81(8_Supplement):349.
[4] Fox PR et al. Circulation. 1995;92:2645-51

Learn More

SCAN ME

SCAN ME

Presented at HFSA 2023 – Clinical Trial Row