EDG-5506 Targets Fast Skeletal Myosin to Protect Dystrophic Muscle and Reduce Muscle Damage Biomarkers in a Phase 1 Trial in Becker Muscular Dystrophy

J Donovan, N Kilburn, G Gordon, B Barthel, M DuVall, A Bronson, A Russell, C Sherman, M Evanchik

Edgewise Therapeutics, Boulder, CO

EDG-5506 Is Designed to Address Root Cause of Muscular Dystrophy by Halting Exaggerated Muscle Damage Due to the Absence of Functional Dystrophin

- Fast (Type II) muscle fibers are affected early and disproportionately in BMD and DMD
- EDG-5506 is a highly selective inhibitor of fast skeletal myosin ATPase, without effects on slow, cardiac, or smooth muscle myosin
- In animal models of DMD, EDG-5506 decreased muscle damage biomarkers, including fast muscle fiber troponin, which is specific to fast muscle fibers. Additionally, fibrosis was decreased, while there were increases in muscle strength and habitual activity

Edgewise THERAPEUTICS 3

EDG-5506 Phase 1 Study Conducted in Healthy Volunteers and Participants with Becker Muscular Dystrophy

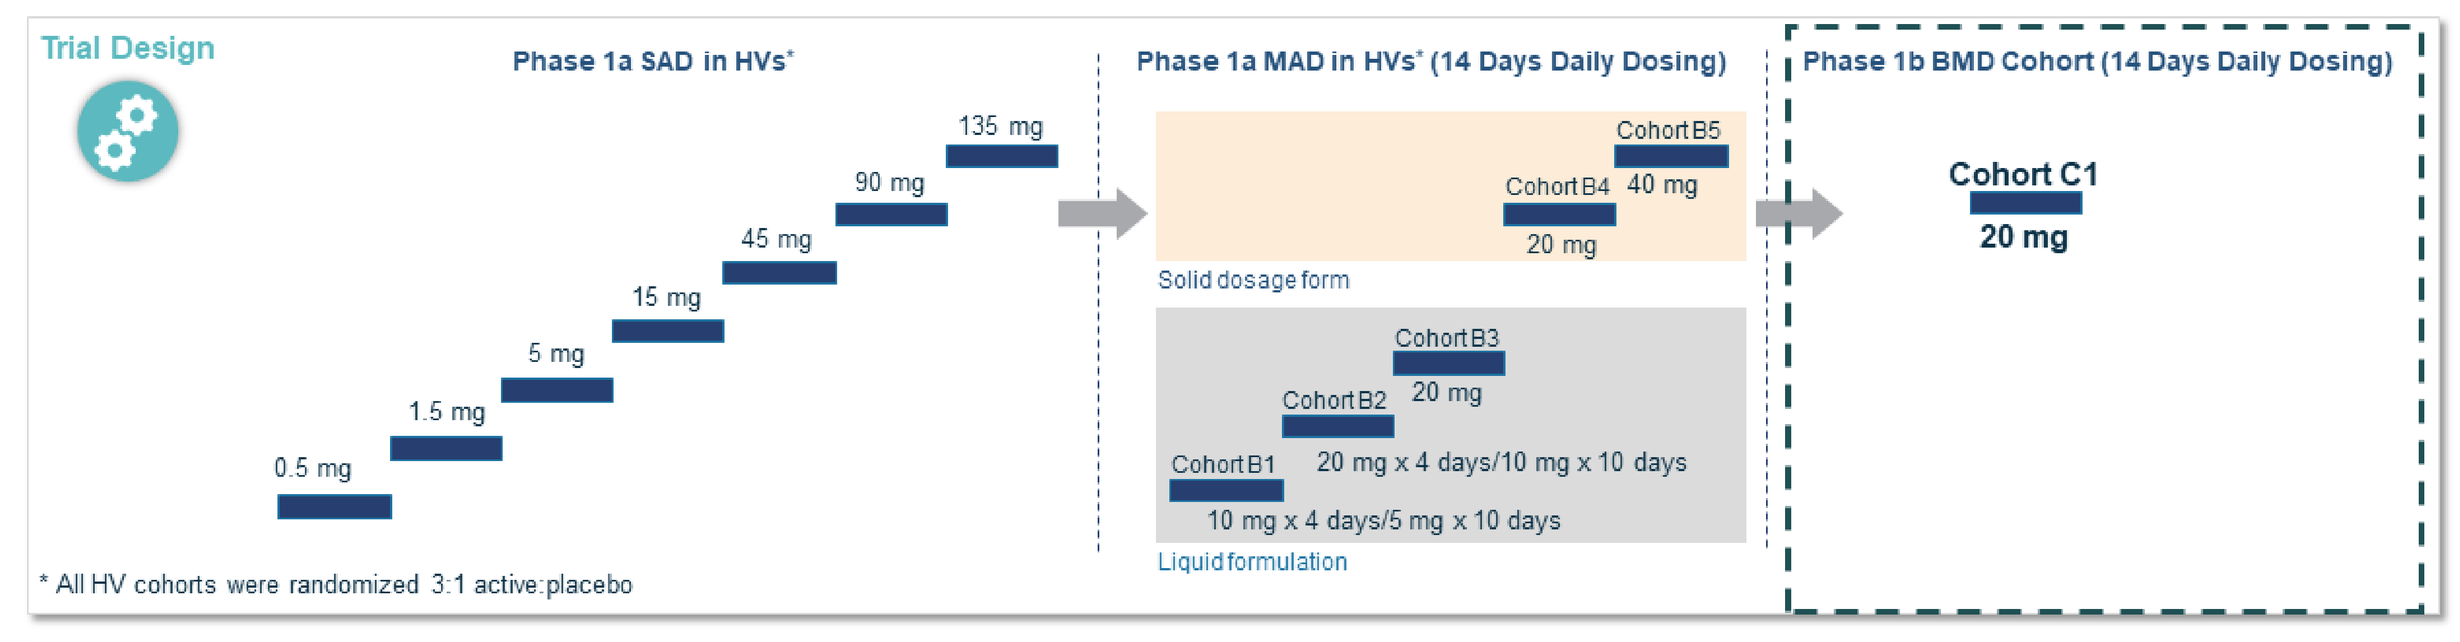
Trial Design
Phase 1a SAD in HVs*
0.5 mg
1.5 mg
5 mg
15 mg
45 mg
90 mg
135 mg
Phase 1a MAD in HVs* (14 Days Daily Dosing)
Cohort B5
Cohort B4 40 mg
20 mg
Solid dosage form
Cohort B3
Cohort B2 20 mg
Cohort B1 20 mg x 4 days/10 mg x 10 days
10 mg x 4 days/5 mg x 10 days
Liquid formulation
Phase 1b BMD Cohort (14 Days Daily Dosing)
Cohort C1
20 mg
* All HV cohorts were randomized 3:1 active:placebo

Key Endpoints

Primary Endpoints
- Safety and tolerability at 20 mg over a 14-day period in BMD

Secondary/Exploratory Endpoints
- Pharmacokinetics, pharmacodynamics
- Assess target tissue engagement judged by muscle/plasma ratio in BMD
- Measurement of serum biomarkers of muscle damage in BMD: CK, fast troponin (TNNI2), myoglobin and SOMAscan, a proteomic panel

Participants were monitored as inpatients for 16 days, with follow-up 1 and 4 weeks after completion of dosing.

Edgewise THERAPEUTICS 4

Overview of Healthy Volunteers (HVs) Phase 1a SAD/MAD with EDG-5506

- EDG-5506 was generally well tolerated with no serious adverse events observed. Most common AEs were somnolence and dizziness, which were generally mild and transient
- Plasma PK showed good oral absorption with or without food, and an extended half-life, consistent with extensive distribution to muscle as was observed preclinically
- EDG-5506 muscle concentrations well above anticipated efficacious levels

Edgewise THERAPEUTICS 5

Participants in the BMD Phase 1b Had Significant Baseline Functional Impairment

Characteristic	BMD Participants (N=7)	Age Normative Values
Age	33.8 years	
Functional Measures (median)		
10-meter walk/run	8.3 sec	< 4 sec
Rise from floor	20 sec	< 3 sec
Serum Creatinine (mean, mg/dL)	0.58	0.92 - 1.16
Serum Creatine Kinase (mean CK, U/L)	1,347	< 205

- Functional tests show significantly compromised or lost function
- Low creatinine consistent with decreased muscle mass
- Elevated CK levels reflect ongoing muscle damage

Edgewise THERAPEUTICS 6

EDG-5506 was Well-Tolerated in BMD Subjects

TEAE	Placebo N=2 n (%)	EDG-5506 (20 mg) N=5 n (%)	Total N=7 n (%)
Any TEAE	**2 (100%)**	**5 (100%)**	**7 (100%)**
Dizziness	2 (100%)	5 (100%)	7 (100%)
Euphoric mood	0	2 (40%)	2 (29%)
Musculoskeletal stiffness	0	2 (40%)	2 (29%)
Somnolence	0	2 (40%)	2 (14%)
Diarrhea	0	1 (20%)	1 (14%)
Nausea	0	1 (20%)	1 (14%)
Fatigue	0	1 (20%)	1 (14%)
Vessel puncture site bruise	0	1 (20%)	1 (14%)
Back pain	0	1 (20%)	1 (14%)
Pain in jaw	0	1 (20%)	1 (14%)
Headache	0	1 (20%)	1 (14%)
Presyncope	0	1 (20%)	1 (14%)
Nasal congestion	0	1 (20%)	1 (14%)
AEs of special interest	**0**	**0**	**0**

- No SAEs
- No discontinuations
- All AEs were mild (Grade 1); AEs were transient and generally declined with increasing exposure

Edgewise THERAPEUTICS 7

No Changes in Grip Strength Observed with Increased EDG-5506 Exposure

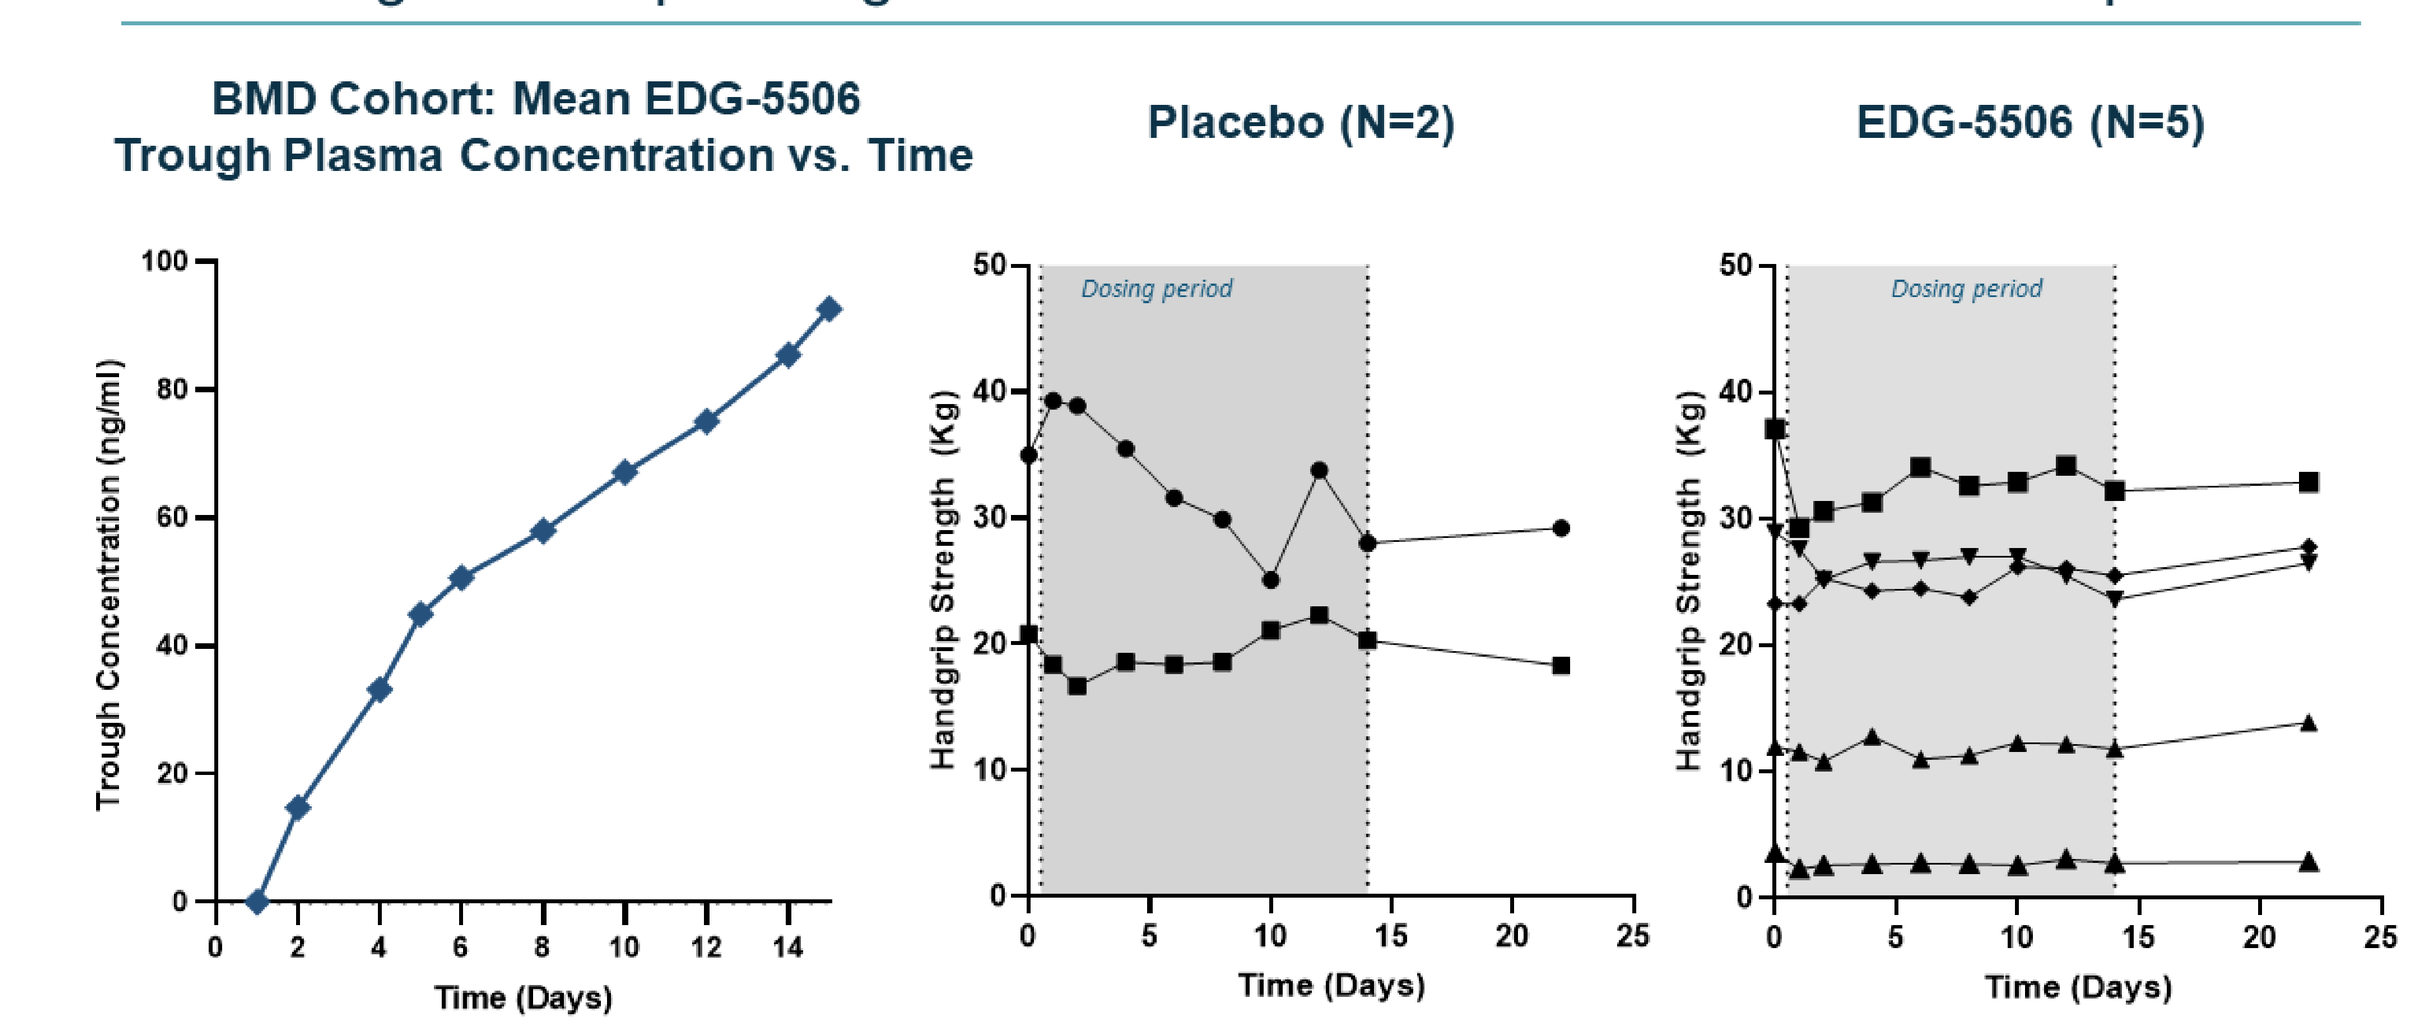
BMD Cohort: Mean EDG-5506 Trough Plasma Concentration vs. Time
Trough Concentration (ng/ml)
Time (Days)
Placebo (N=2)
EDG-5506 (N=5)
Dosing period
Handgrip Strength (Kg)
Time (Days)

Edgewise THERAPEUTICS 8

EDG-5506 Concentrates in Dystrophic Muscle Above Levels Predicted to Provide Meaningful Clinical Benefit

Formulation	Cohort		Maintenance Dose (mg)	Day 14 Tissue Levels* Muscle (ng/g)
Liquid Formulation	HVs	B1	5	980
		B2	10	2,740
		B3	20	4,360
Solid Dosage Form		B4	20	6,140
		B5	40	6,570
	BMD	C1	20	5,155†

** In HVs concentrations after 14 days are estimated to be half of steady state*
† VL biopsy levels adjusted for ~60% fat fraction in BMD subjects

Target human muscle exposure range: 1,000-4,100 ng/g

Edgewise THERAPEUTICS 9

Key Biomarkers of Muscle Damage Significantly Decreased with EDG-5506

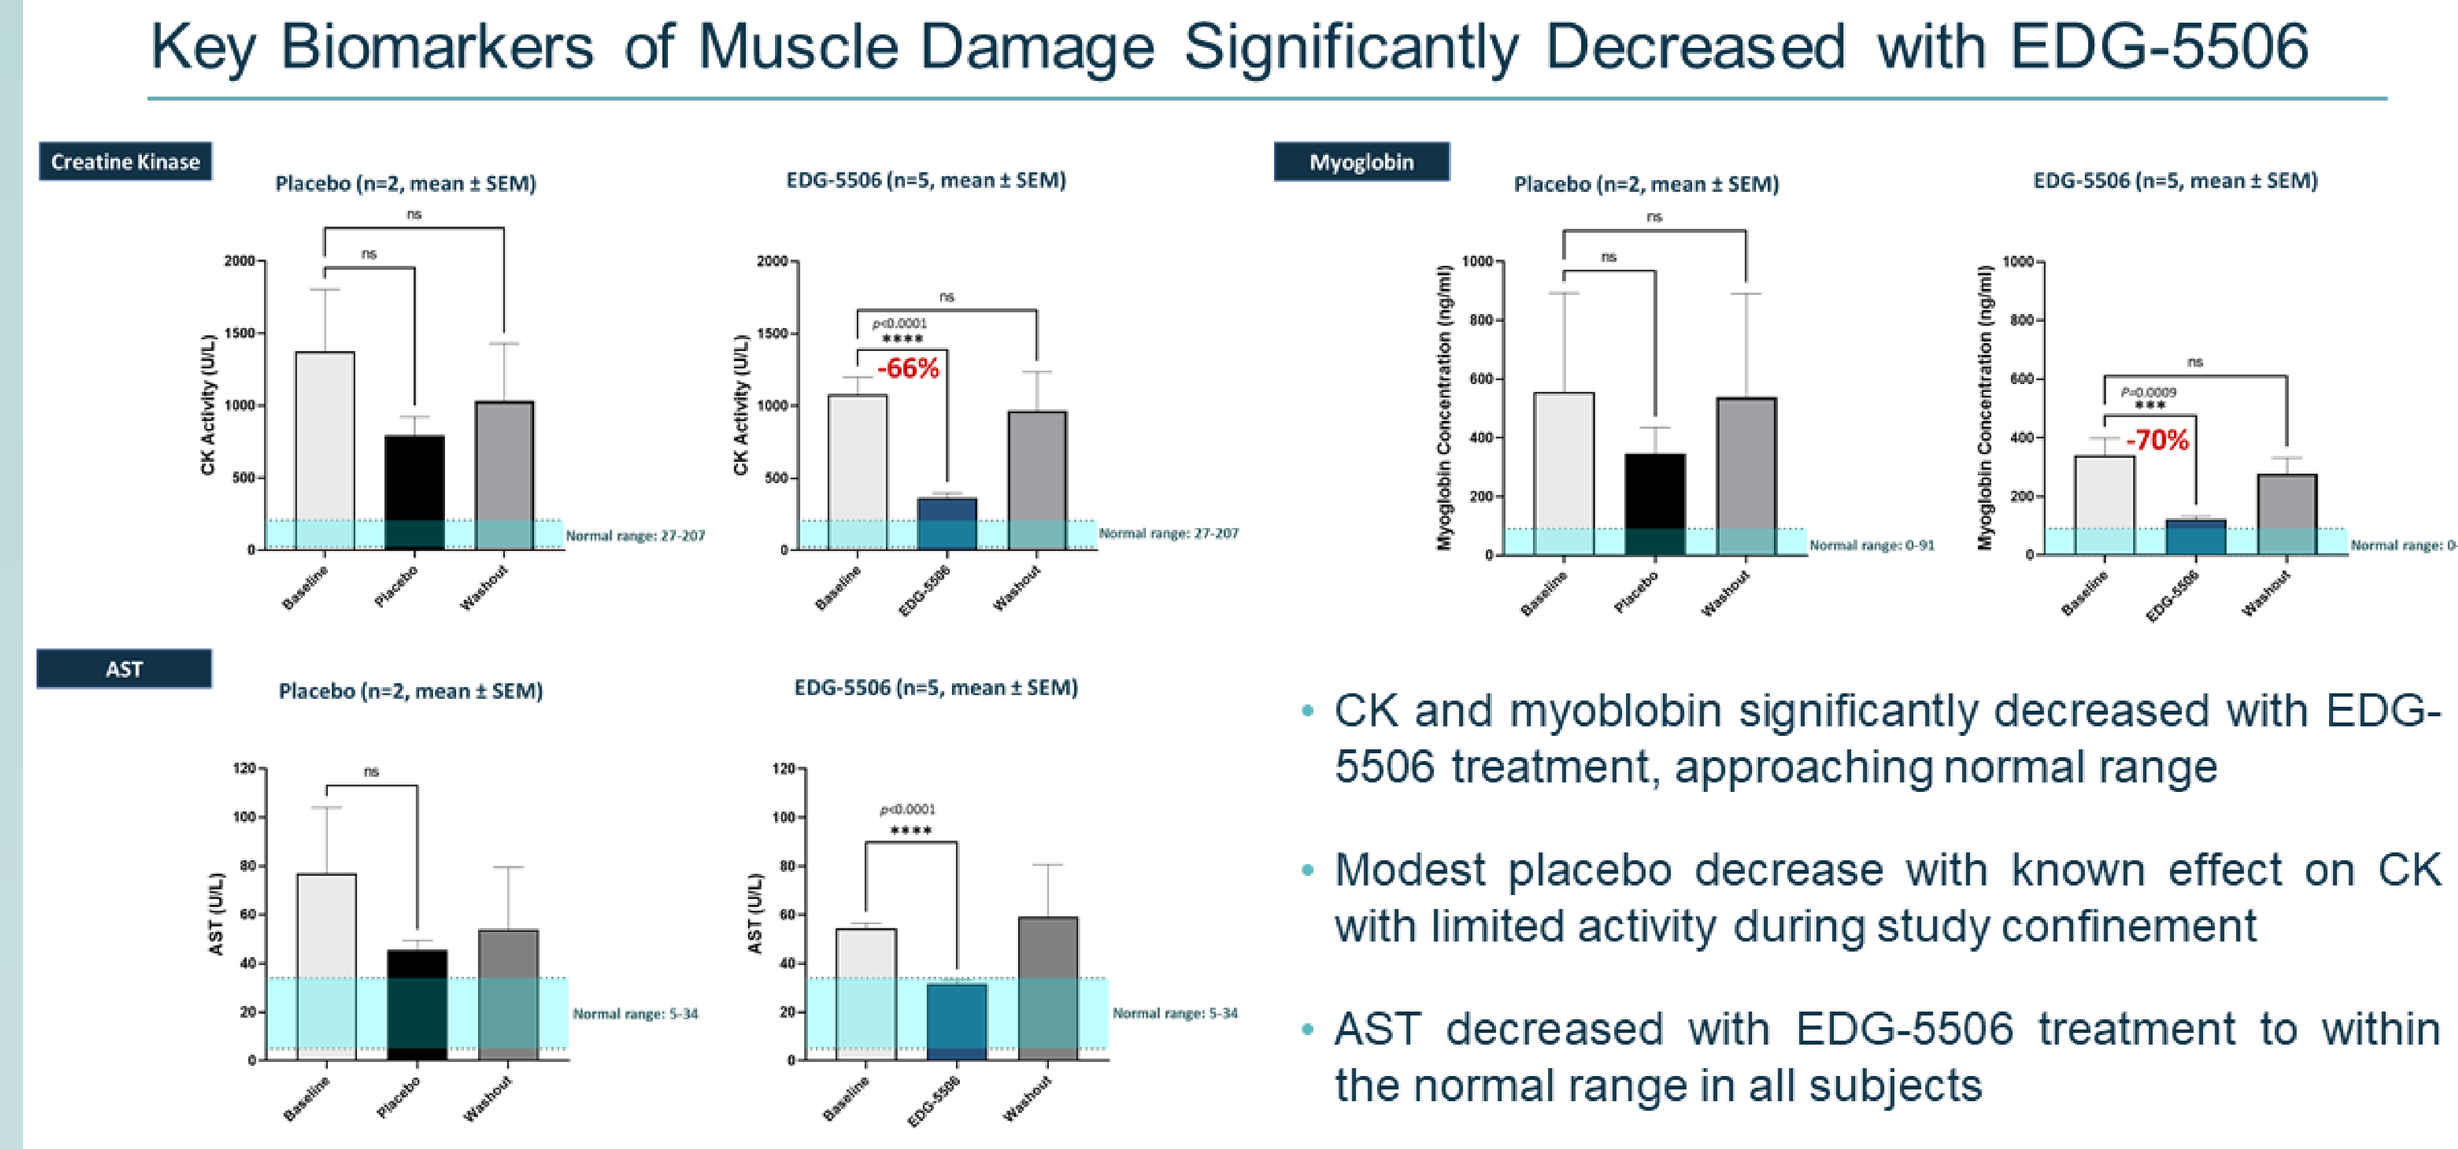
Creatine Kinase
Placebo (n=2, mean ± SEM)
EDG-5506 (n=5, mean ± SEM)
-66%
Myoglobin
Placebo (n=2, mean ± SEM)
EDG-5506 (n=5, mean ± SEM)
-70%
AST
Placebo (n=2, mean ± SEM)
EDG-5506 (n=5, mean ± SEM)

- CK and myoblobin significantly decreased with EDG-5506 treatment, approaching normal range
- Modest placebo decrease with known effect on CK with limited activity during study confinement
- AST decreased with EDG-5506 treatment to within the normal range in all subjects

Edgewise THERAPEUTICS 10

Using SOMAscan 7,000 Analyte Set, A Proteomic Signature for BMD Was Identified

Baseline BMD vs. Healthy Biomarker Fingerprint Analysis

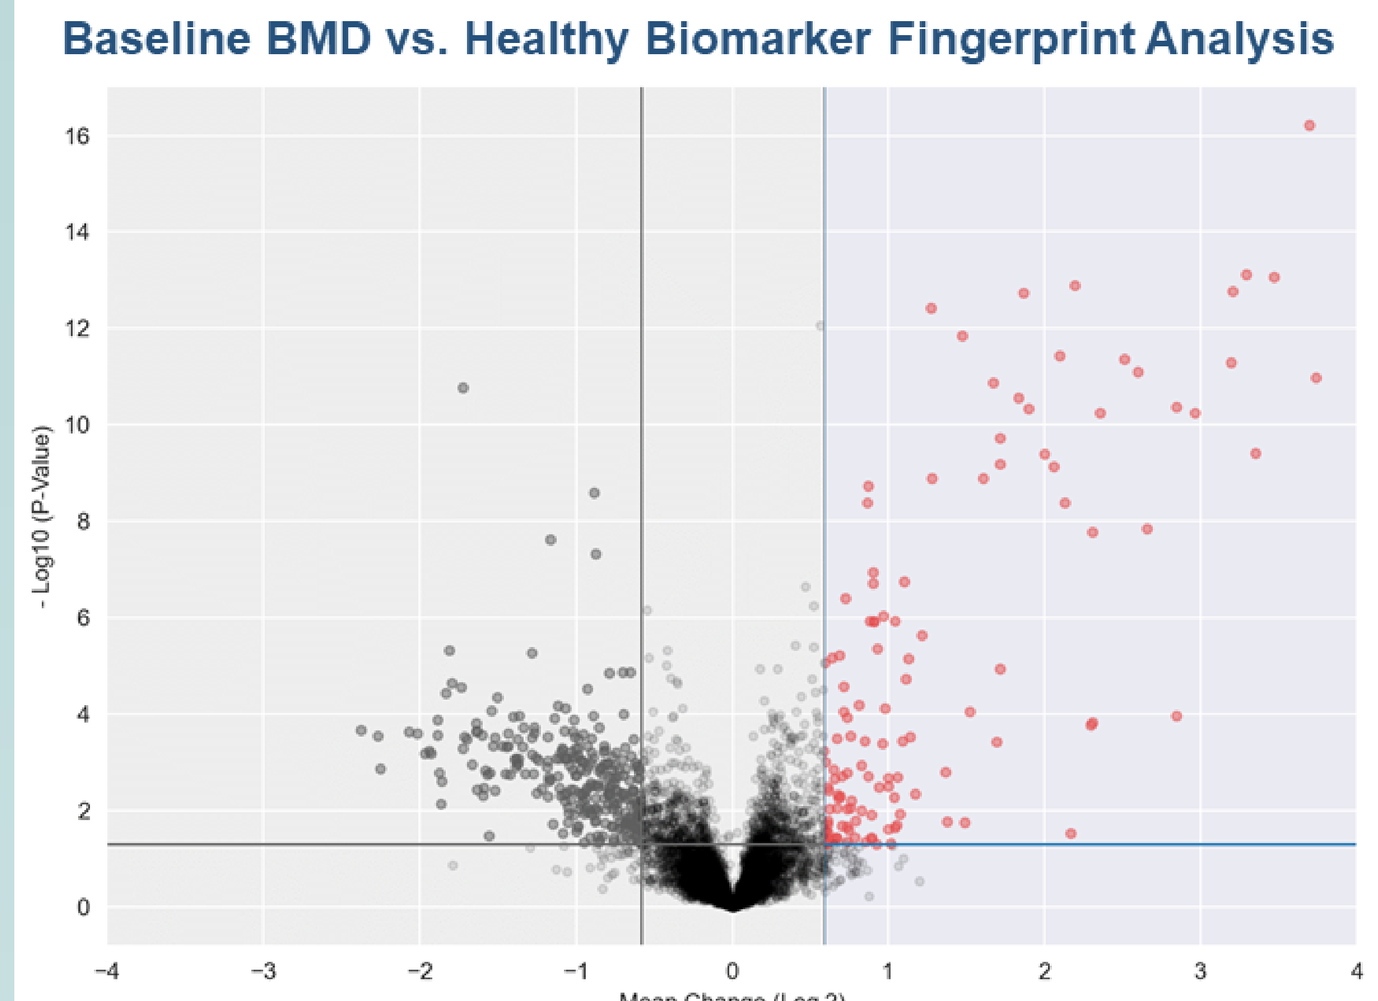
- Log10 (P-Value)
Mean Change (Log 2)

- SOMAscan allows unbiased protein profiling of elevated plasma markers in BMD
- Baseline plasma samples (n=7) were compared to baseline samples taken from HVs (n=25)
- Proteins filtered by magnitude of change (≥1.5X) and adjusted *p* value (<0.05) vs. HV
- Baseline analysis identified a fingerprint of elevated proteins in BMD
- Most significant proteins are from muscle and metabolic pathways consistent with muscle injury

Edgewise THERAPEUTICS 11

Robust, Significant and Time-Dependent Decreases in Elevated BMD Biomarkers

Changes in BMD biomarkers vs. placebo* (Day 14) | **BMD biomarkers responsive to increased exposure to EDG-5506**

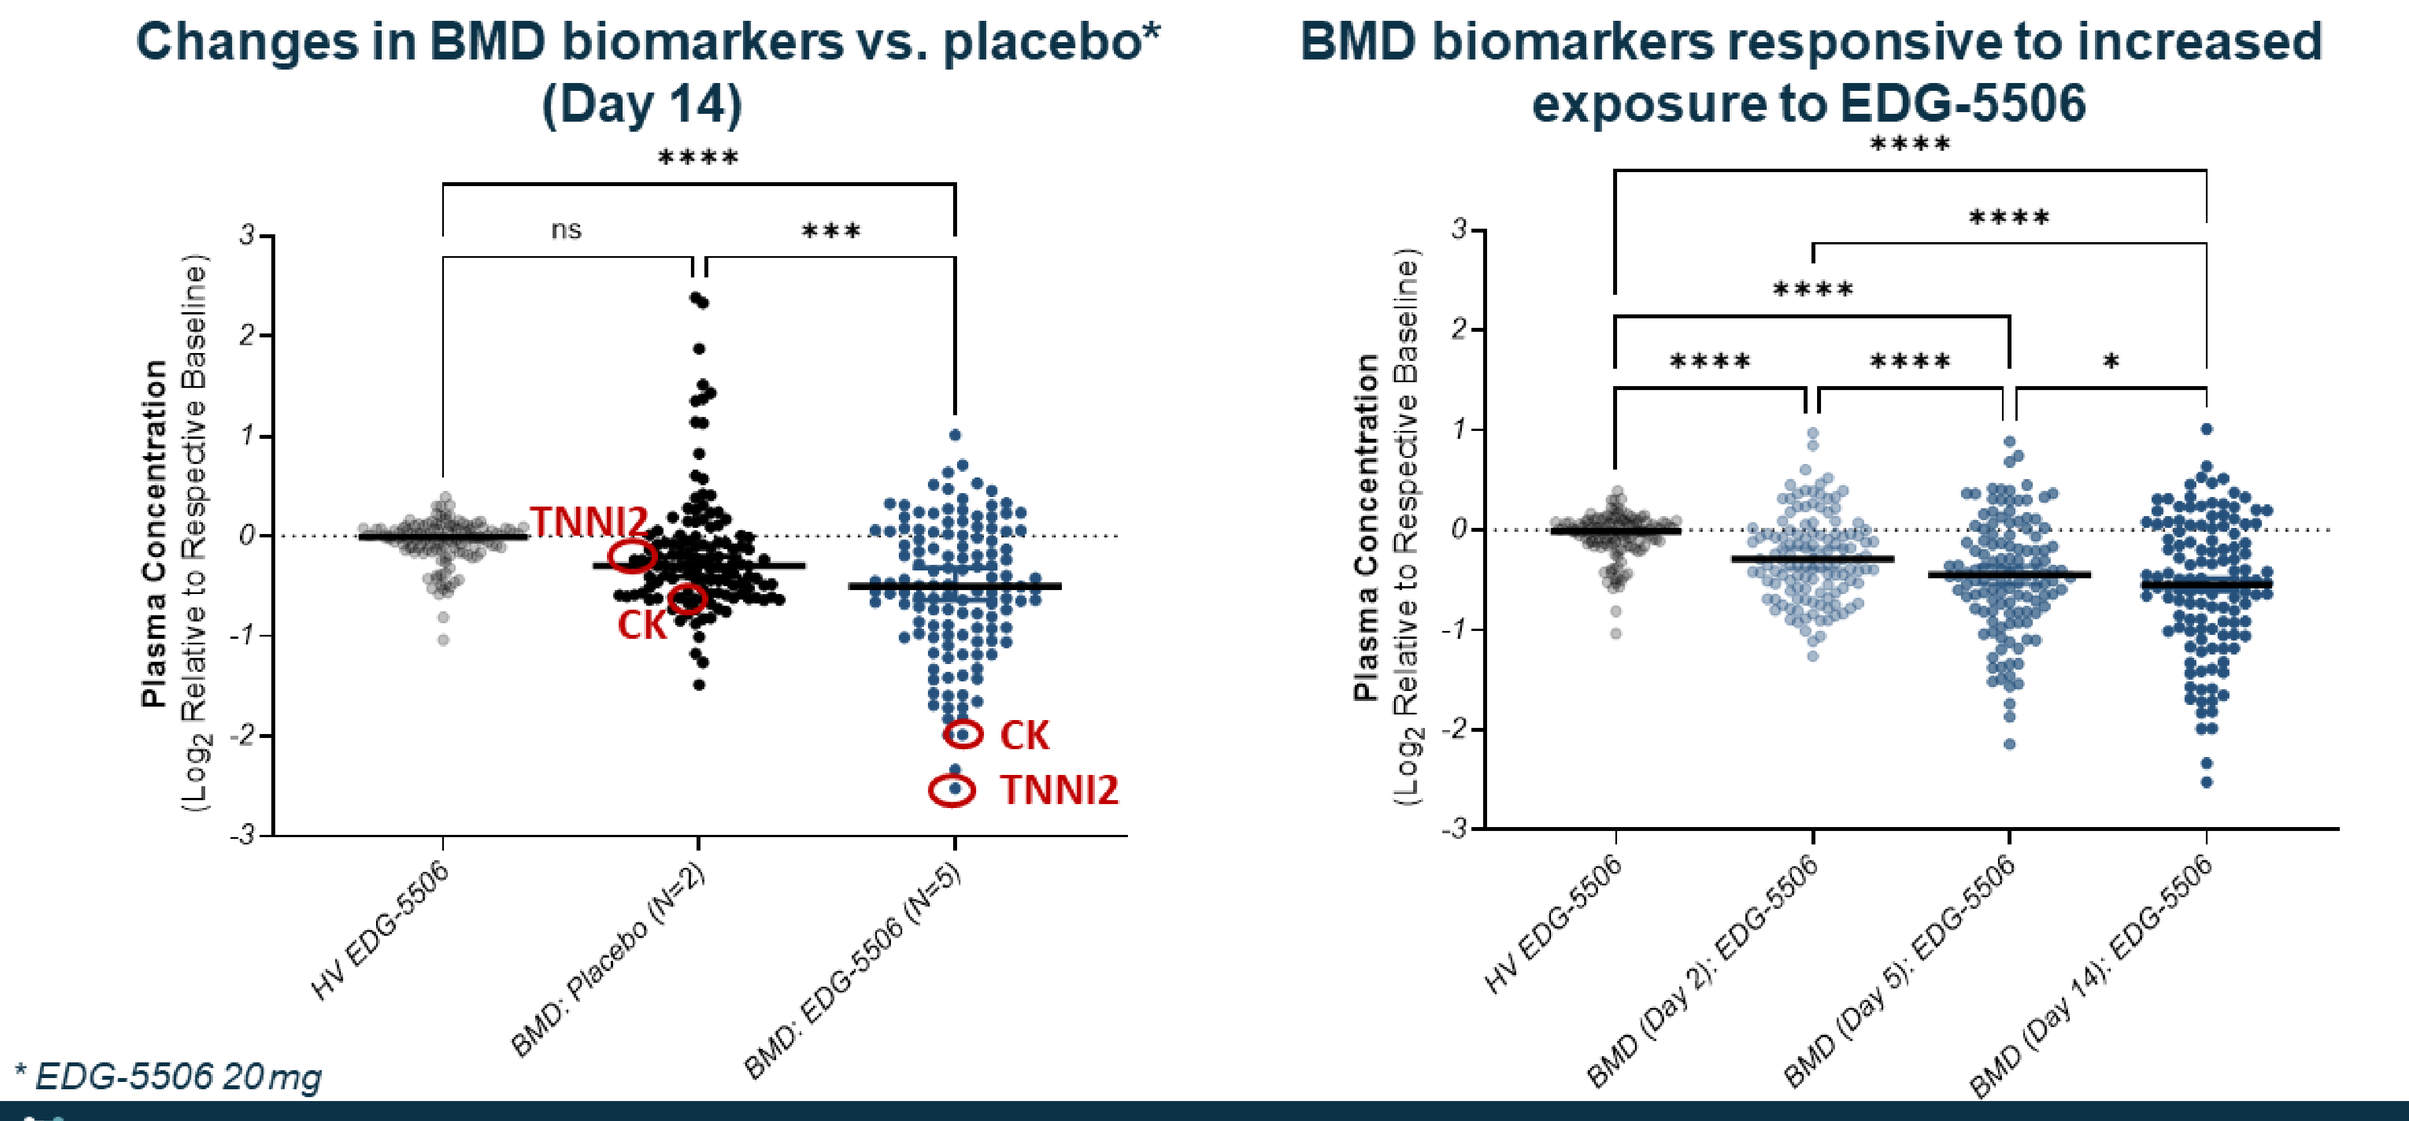
Plasma Concentration
(Log2 Relative to Respective Baseline)
TNNI2
CK
CK
TNNI2
HV EDG-5506
BMD: Placebo (N=2)
BMD: EDG-5506 (N=5)
BMD (Day 2): EDG-5506
BMD (Day 5): EDG-5506
BMD (Day 14): EDG-5506

** EDG-5506 20 mg*

Edgewise THERAPEUTICS 12

With SOMAscan CK and Fast Troponin Reduced to Levels Near Those Observed in HVs Following Treatment with EDG-5506

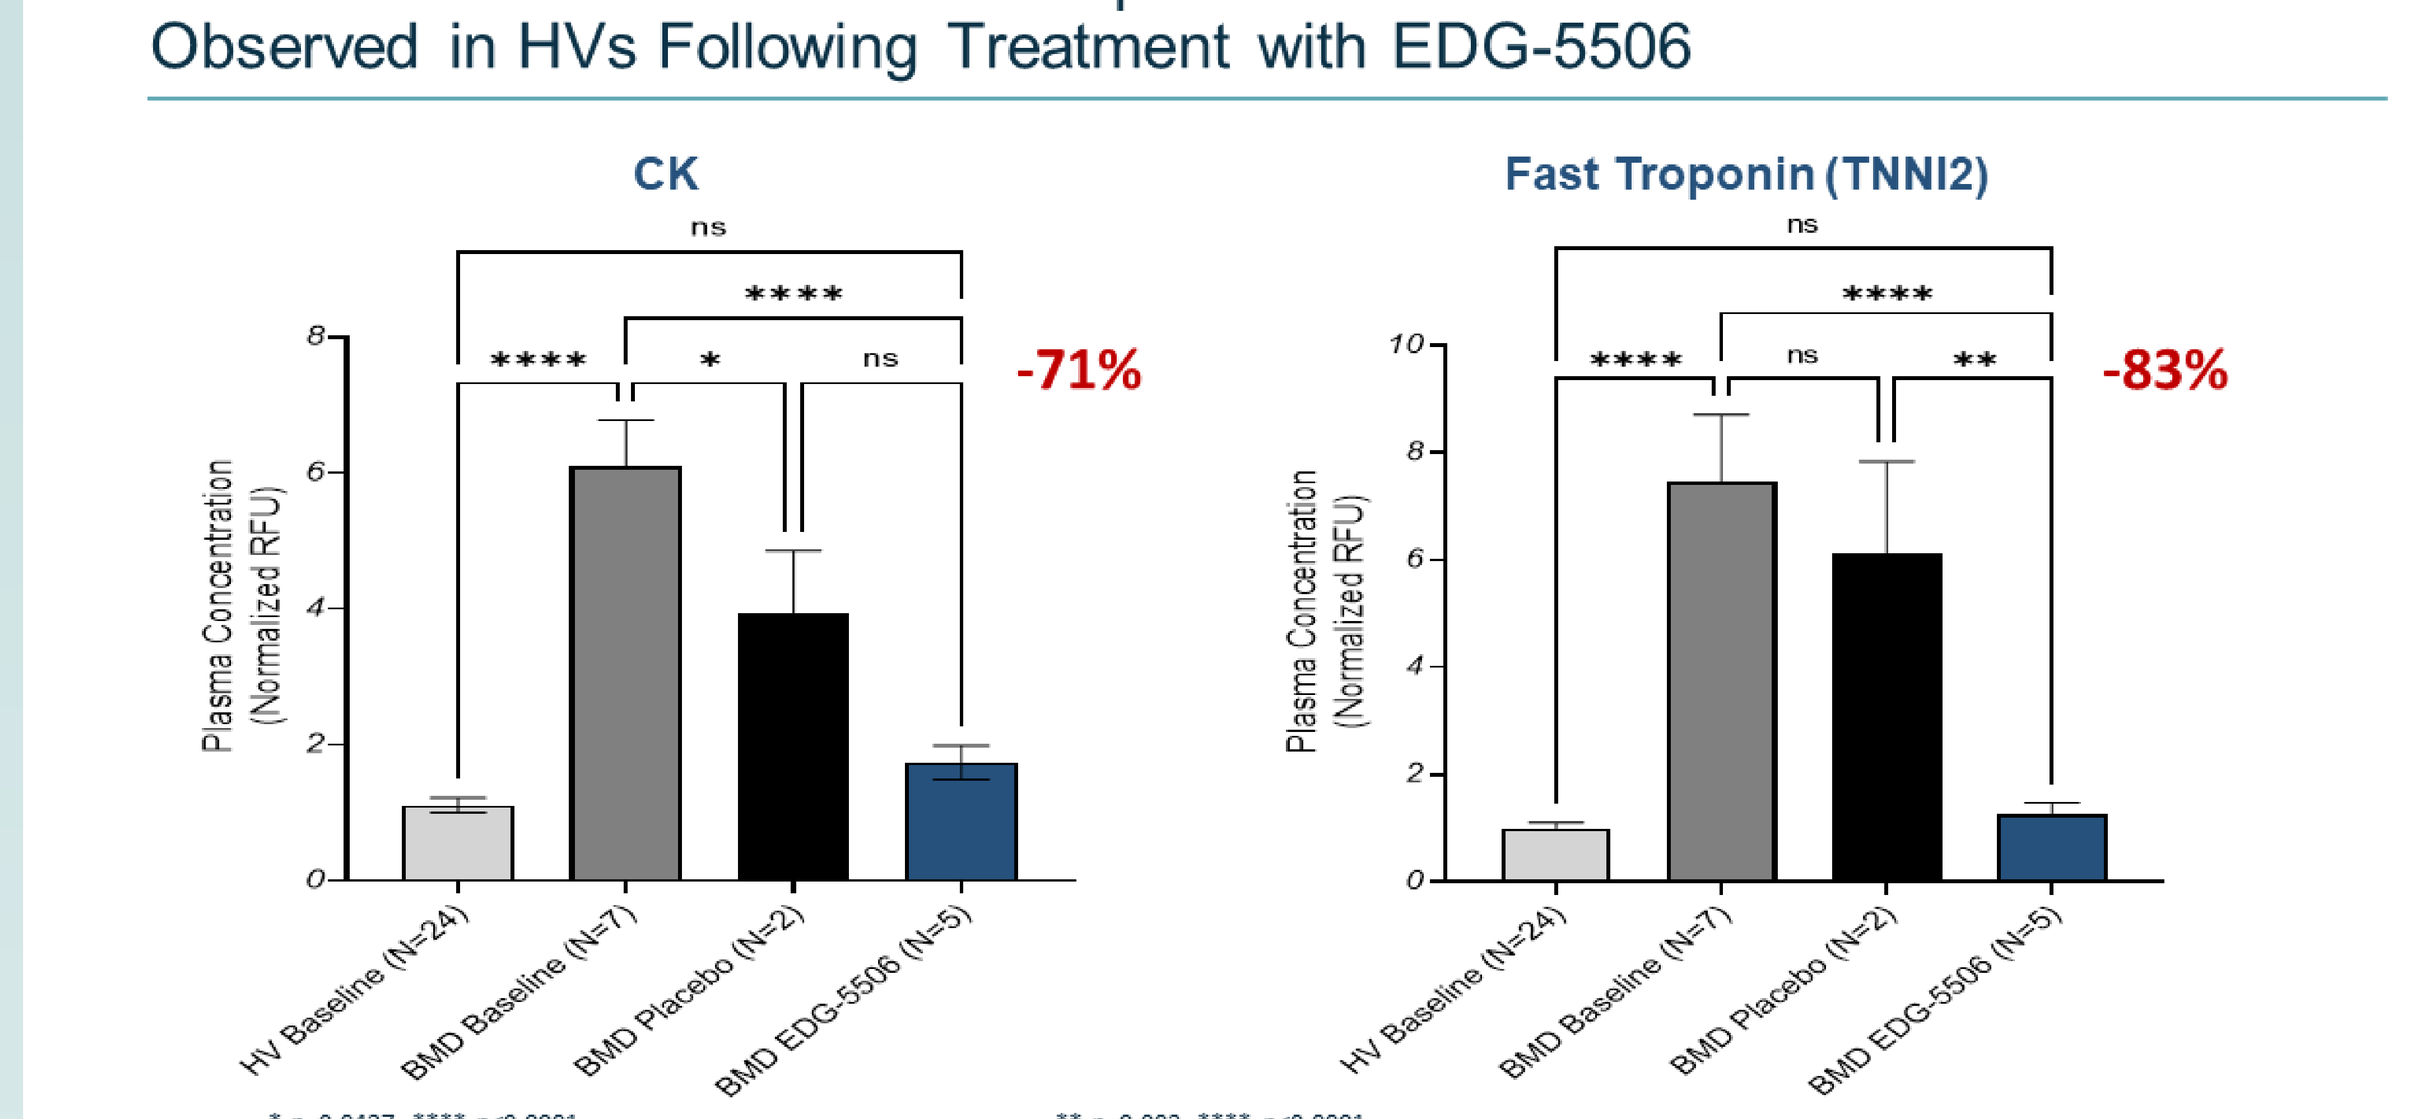
CK
-71%
Fast Troponin (TNNI2)
-83%
Plasma Concentration
(Normalized RFU)
HV Baseline (N=24)
BMD Baseline (N=7)
BMD Placebo (N=2)
BMD EDG-5506 (N=5)
* p=0.0427, **** p<0.0001
** p=0.002, **** p<0.0001

Mean ± SEM

Edgewise THERAPEUTICS 13

Consistent Muscle Injury Proteomic Response with EDG-5506 but not Placebo

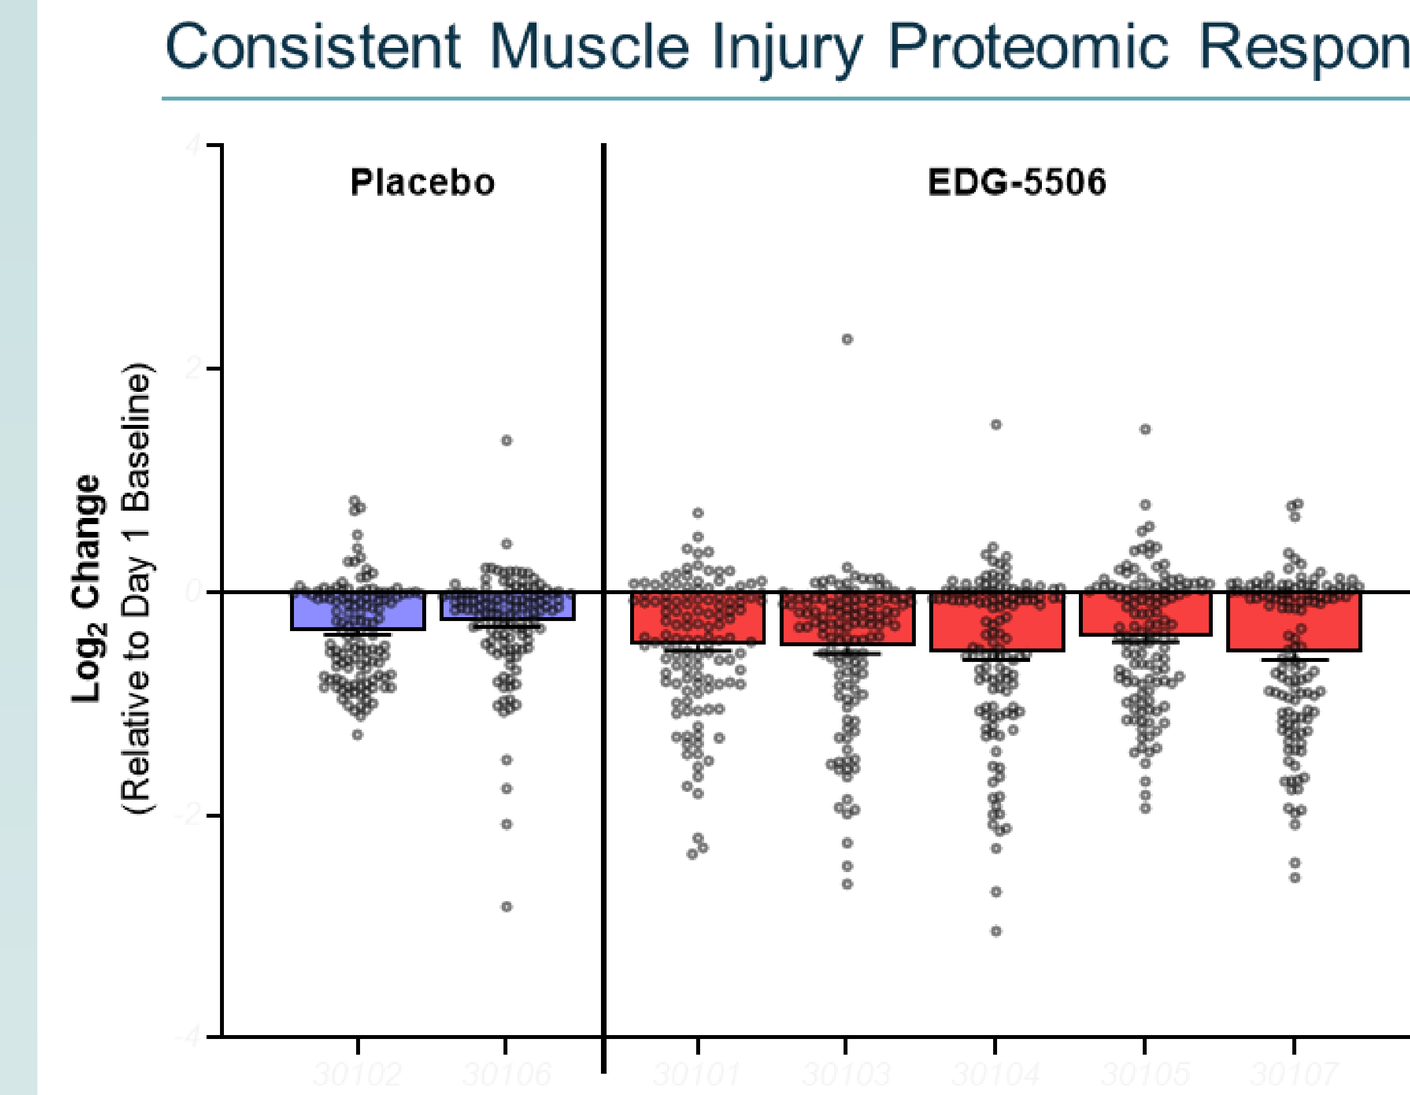
Placebo
EDG-5506
Log2 Change
(Relative to Day 1 Baseline)
Individual Participants

Proteins with greatest reductions	PBO		EDG-5506				
Fast troponin (TNNI2)*	-	-	X	X	X	X	X
Myosin reg light chain 2	-	-	-	X	X	X	X
MYBP-C*	-	-	X	-	X	-	-

*specific to fast muscle fibers

Edgewise THERAPEUTICS 14

Consistent and Progressive EDG-5506 Effect on Exercise Responsive Markers

- In adults with BMD we used SOMAscan samples from an exercise study to define a proteomic signature that was elevated compared to controls at baseline, and had an exaggerated increase with exercise (Poster # 155)
- These proteins, largely characterized by enrichment in muscle, rapidly and progressively decreased with EDG-5506 but not with placebo

EDG-5506 (N=5)

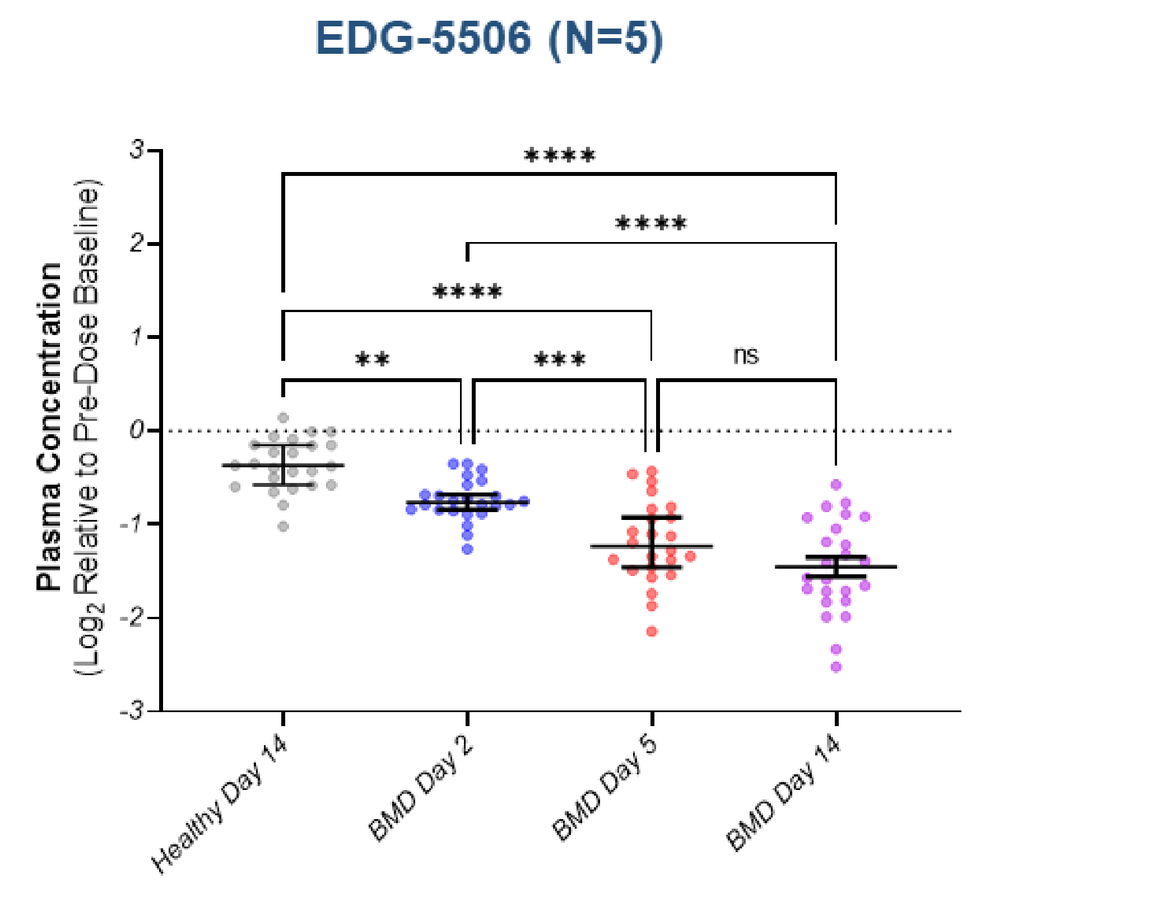
Plasma Concentration
(Log2 Relative to Pre-Dose Baseline)
Healthy Day 14
BMD Day 2
BMD Day 5
BMD Day 14

Rapid and progressive decrease

Edgewise THERAPEUTICS 15

Elevated Biomarkers Decrease Most Following EDG-5506

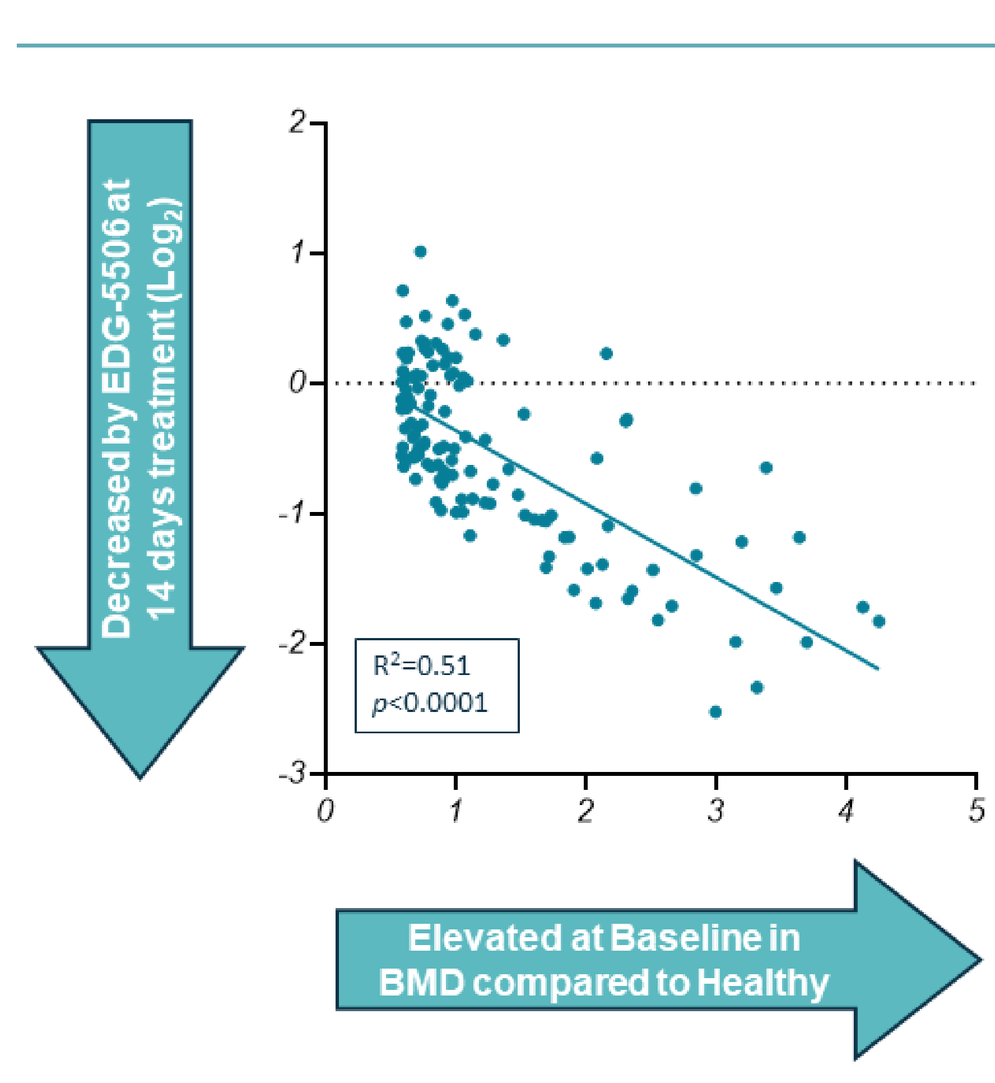
Decreased by EDG-5506 at 14 days treatment (Log2)
R²=0.51
p<0.0001
Elevated at Baseline in BMD compared to Healthy

- Close association of elevated BMD biomarkers with EDG-5506 response reveals a significant relationship to biomarker lowering
- Overall, this suggests a broad normalization of the BMD proteomic signature rather than a change in just a subset of biomarkers

Edgewise THERAPEUTICS 16

EDG-5506: Well-Tolerated with Decreases in Biomarkers of Muscle Damage in BMD Subjects

- EDG-5506 was well tolerated; adverse events were transient, mild and similar to those observed in HVs
- Plasma and muscle exposure levels at or above efficacious levels in animal models
- Rapid, significant reduction in multiple biomarkers of muscle damage observed after only 2 weeks of dosing
- Preventing muscle damage is anticipated to provide benefit to individuals with dystrophinopathy, both BMD and DMD
- Phase 2 studies in BMD and DMD are planned

Edgewise THERAPEUTICS 17

Acknowledgments

The authors are grateful to the participants in the trial

Disclaimer

EDG-5506 is an investigational drug that is not approved in any territory.
The authors are employees or consultants for Edgewise Therapeutics and may hold stock and/or stock options

Edgewise THERAPEUTICS

Presented at the MDA Clinical and Scientific meeting, Nashville TN, March 2022